Development of screening in CD4+ T cells to identify drug targets for immune-mediated inflammatory diseases

Giorgia Bergamin, Ricky Trigg, Lewis Brayshaw, Becky Saunders, Rob Jepras, Lorna Suckling, Maria New, Richard Kasprowicz, Wilson Li, Ketil Tvermosegaard, David Tough, Rebecca Randle, Huw Lewis, and Klio Maratou.

Abstract

Immune-mediated inflammatory diseases (IMIDs) are a group of disorders characterised by tissue inflammation as a result of dysregulated immune responses. Enhancing the production of anti-inflammatory cytokines from T cells may balance the inflammatory state of IMIDs and provide therapeutic benefit. To this end, we developed a genome-wide CRISPR screen in primary CD4+ T cells to identify genes which, when knocked out, lead to increased release of an anti-inflammatory cytokine. We extensively optimised several elements of the screening workflow including lentiviral transduction, lentivirus production, Cas9 electroporation in bulk cell populations and FACS sorting, and ran a focused screen based on a mini library of ~1,000 gRNAs to assess the performance of the workflow before conducting a genome-wide screen.

Aim

Develop and execute a pooled, CRISPR screening workflow in primary CD4+ T cells to investigate genetic targets which, when knocked out, lead to upregulation of an anti-inflammatory cytokine.

Introduction

IMID is an umbrella term for a group of autoimmune inflammatory disorders characterised by excessive tissue inflammation and uncontrolled production of antibodies. These disorders are driven by an imbalance of immunoregulatory cytokines due to a predominant T_h1 lymphocyte response (Figure 1). A potential therapeutic approach for IMIDs is to reverse the inflammatory state by upregulating release of anti-inflammatory cytokines from resident T cells in affected tissues. CRISPR technologies enable us to investigate the genes which play a critical role in the production of anti-inflammatory cytokines, and lead to the identification of novel drug targets for IMIDs.

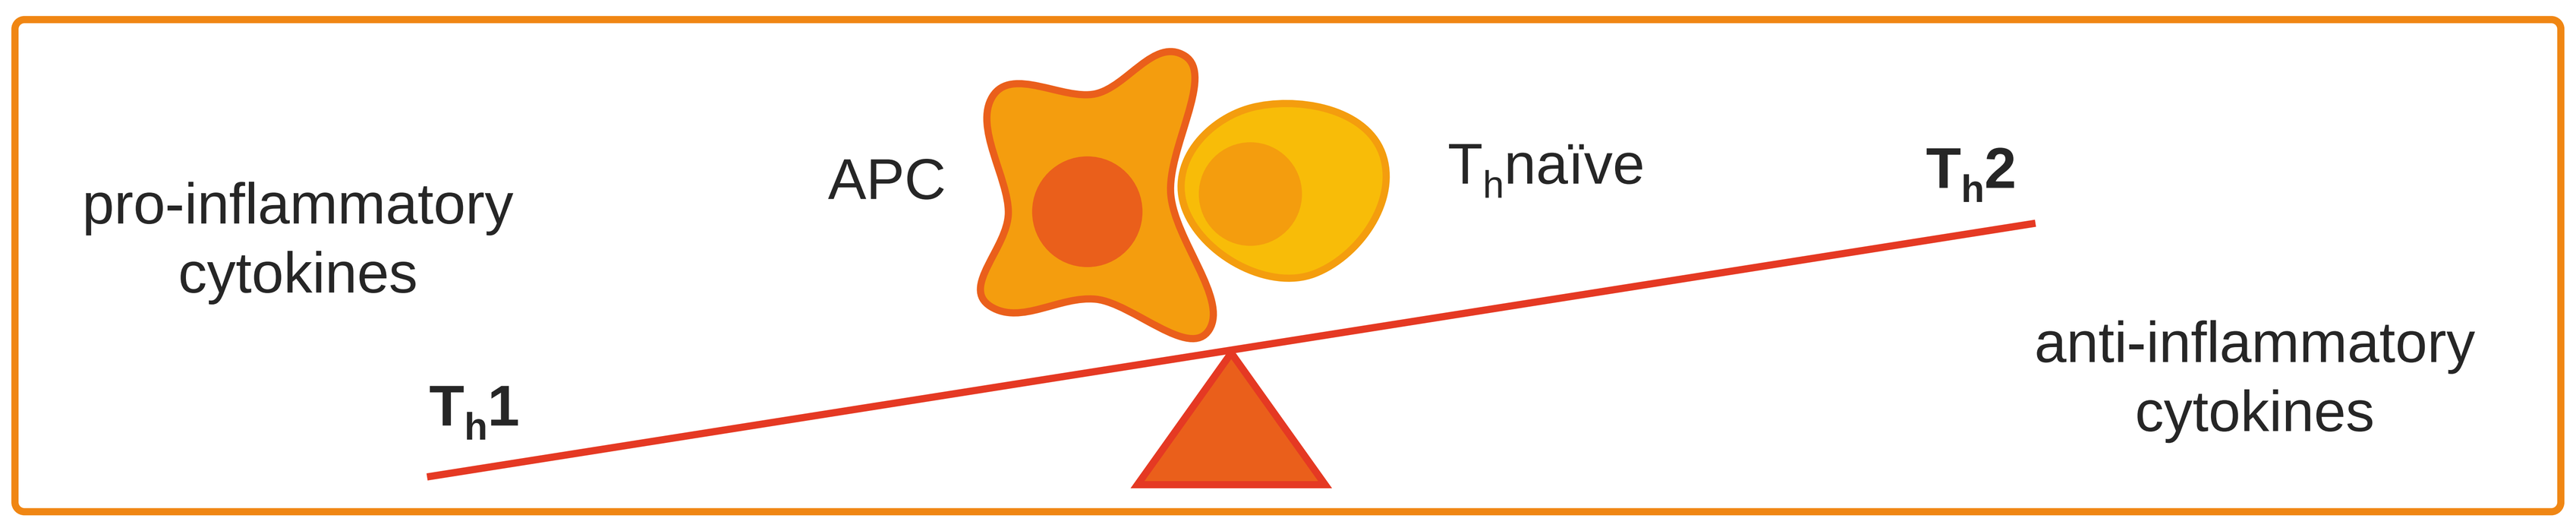
pro-inflammatory cytokines
APC
T_hnaïve
T_h2
anti-inflammatory cytokines
T_h1

Fig 1. Immune-mediated inflammation results from a predominant T_h1 response

Results I – Development of pooled CRISPR screen

The human biological samples in this study were sourced ethically and their research use was in accord with the terms of the informed consents under an IRB/EC approved protocol.

Optimisation of SLICE workflow

To perform the pooled CRISPR screen in primary T cells, we adapted the SLICE protocol (Shifrut et al., Cell. 2018;175(7):1958–1971). This method overcomes the problem of intolerance of constitutive Cas9 expression by primary T cells, via delivery in protein format through electroporation. Several steps of the protocol required optimisation (Fig 2).

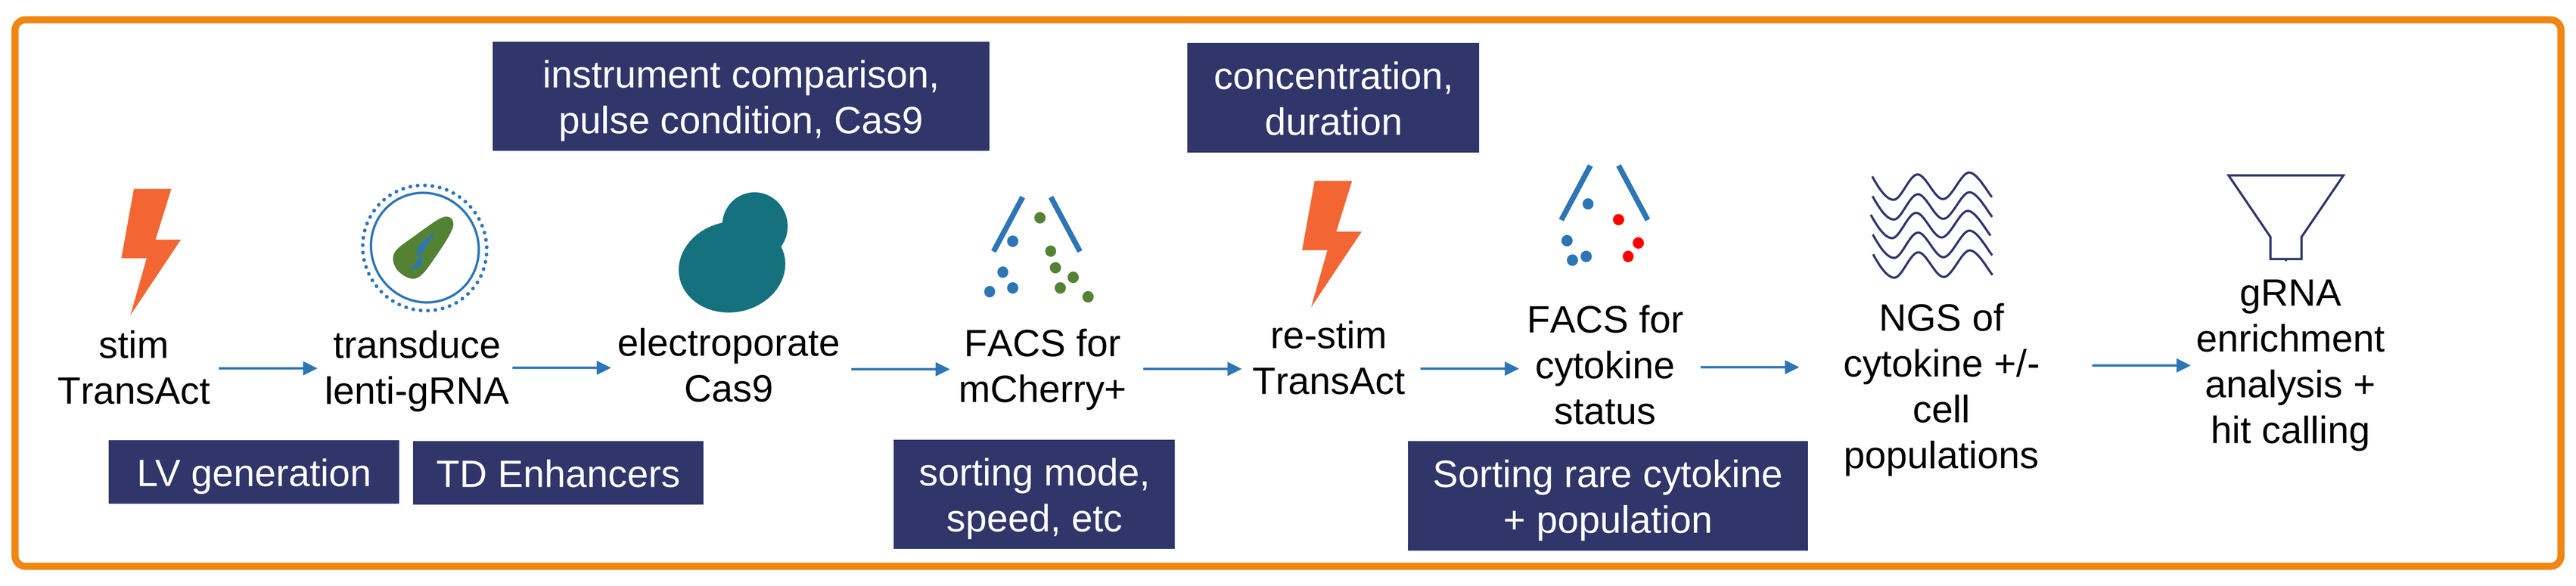
instrument comparison, pulse condition, Cas9
concentration, duration
stim TransAct
transduce lenti-gRNA
electroporate Cas9
FACS for mCherry+
re-stim TransAct
FACS for cytokine status
NGS of cytokine +/- cell populations
gRNA enrichment analysis + hit calling
LV generation
TD Enhancers
sorting mode, speed, etc
Sorting rare cytokine + population

Fig 2. Overview of SLICE workflow and key areas for optimisation

Transduction efficiency: boosting lentivirus production using ViralBoost reagent (Alstem) (Fig 3A) and coating wells before transduction using RetroNectin® (Takara) (Fig 3B) significantly improves transduction efficiency of primary human T-cells.

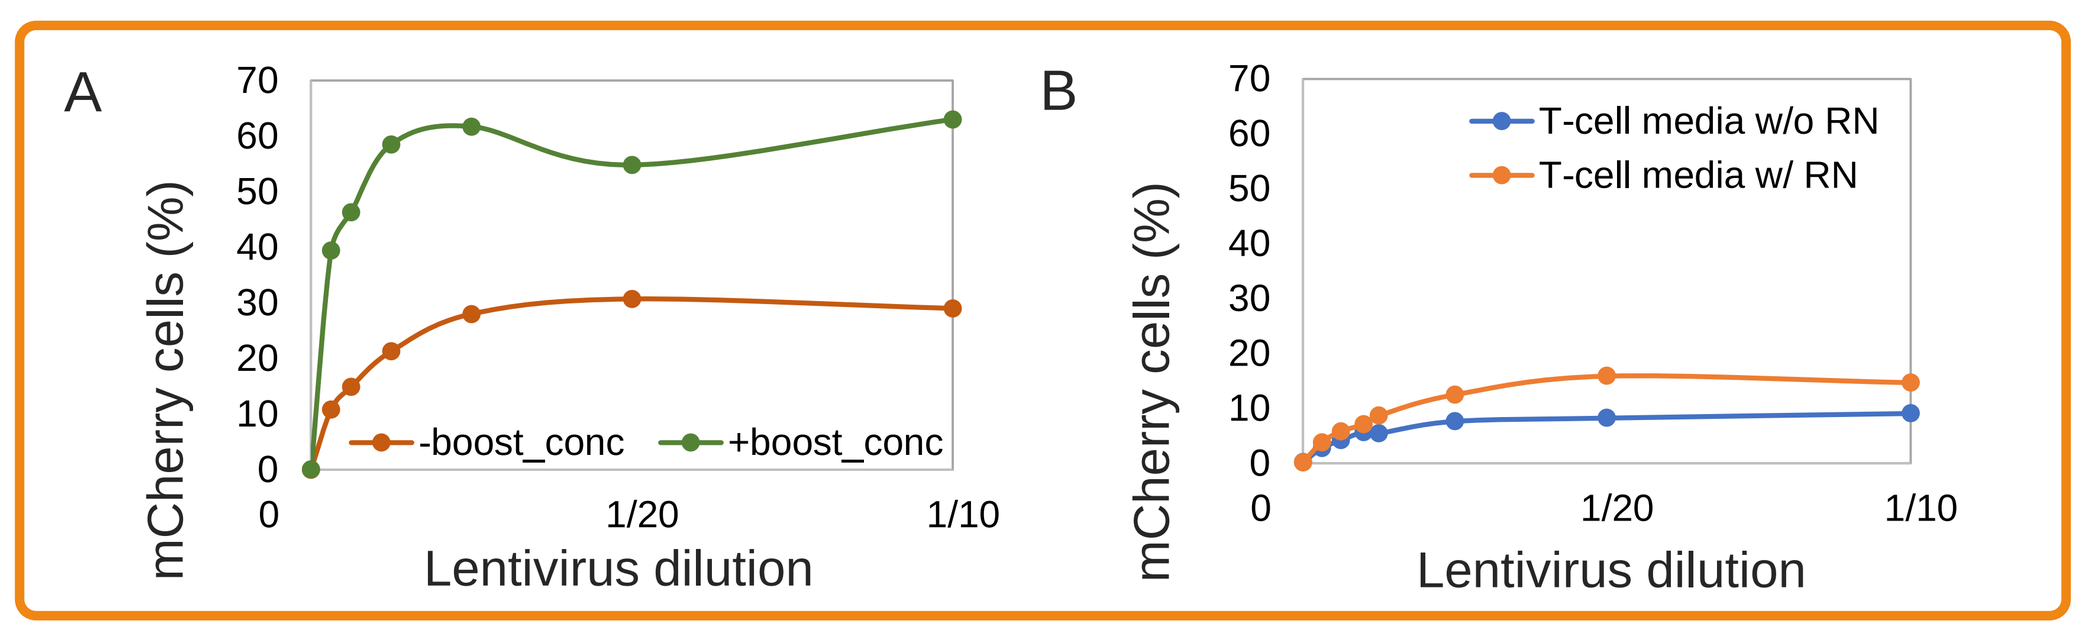
A
mCherry cells (%)
-boost_conc
+boost_conc
Lentivirus dilution
B
T-cell media w/o RN
T-cell media w/ RN
Lentivirus dilution

Fig 3. Enhancing transduction efficiency

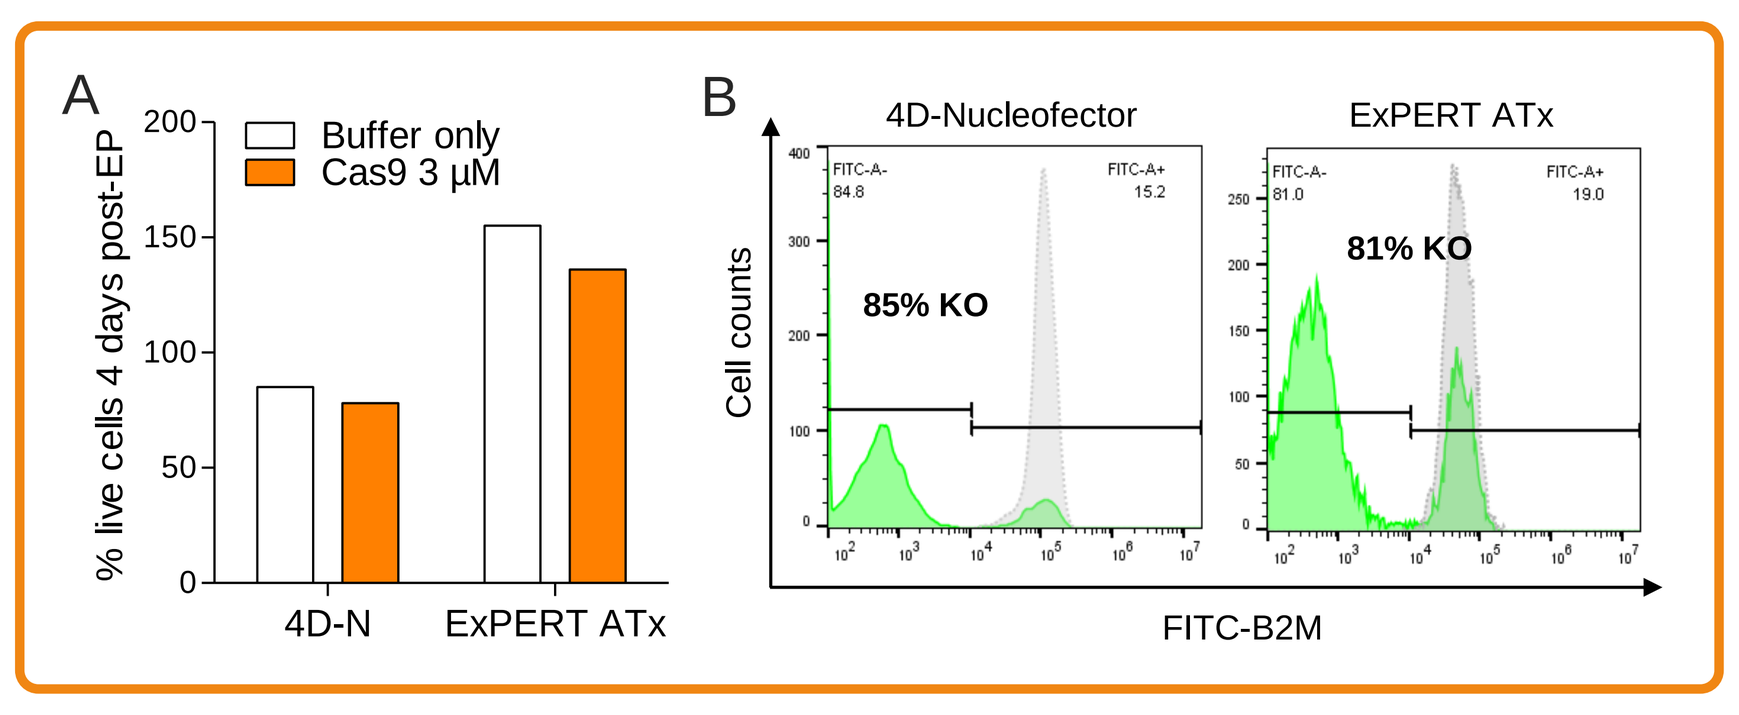
A
% live cells 4 days post-EP
Buffer only
Cas9 3 µM
4D-N
ExPERT ATx
B
4D-Nucleofector
ExPERT ATx
85% KO
81% KO
Cell counts
FITC-B2M

Fig 4. Comparison of cell viability and B2M KO efficiency between 4D-Nucleofector and EXPERT ATx

Cas9 electroporation: electroporation of Cas9 into cells transduced with sgRNA for B2M using the MaxCyte ExPERT ATx ('Expanded T-cell 2' program) improves viability of cells after EP compared to the Lonza 4D-Nucleofector (EH115 program) (Fig 4A) while maintaining the KO efficiency as tested via flow cytometry (Fig 4B).

mCherry sorting: sorting cells at 20,000 evt/sec in yield mode using FACS Aria Fusion with 70µm nozzle allows to enrich mCherry+ cells without affecting viability and cell growth post sorting (Fig 5A). Sorting is preferable to antibiotic selection, as it is faster and does not affect the ability of cells to respond to the secondary stimulation. Representative plots of mCherry cells pre- and post-sort are shown in Fig 5B.

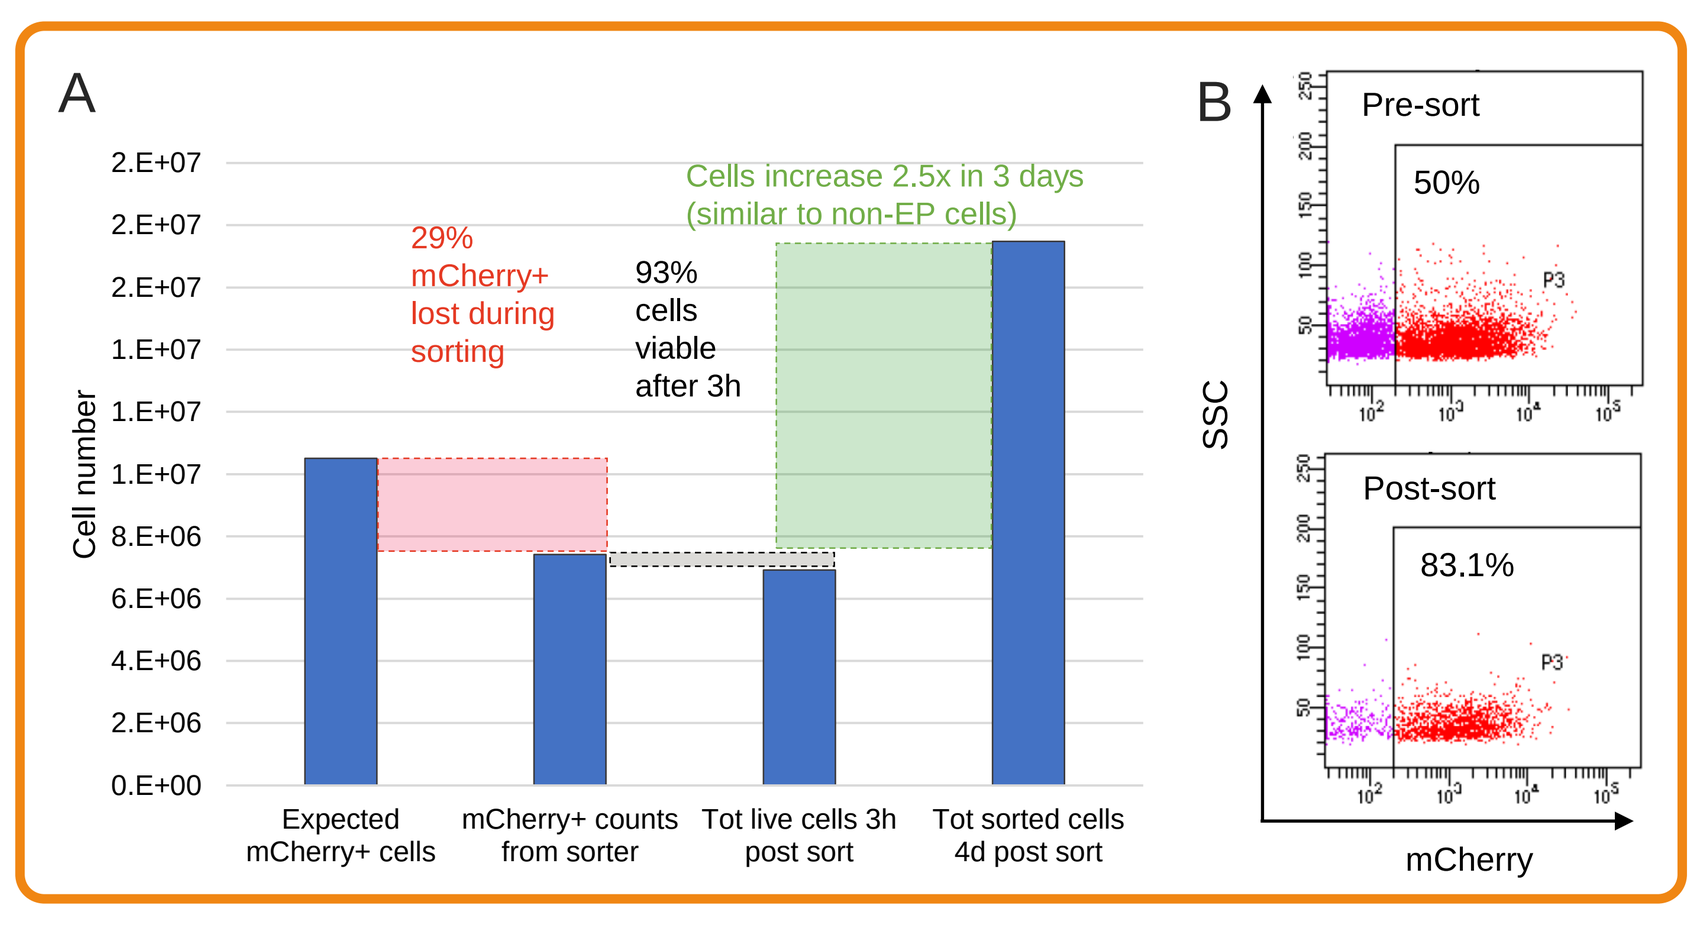
A
Cell number
29% mCherry+ lost during sorting
93% cells viable after 3h
Cells increase 2.5x in 3 days (similar to non-EP cells)
Expected mCherry+ cells
mCherry+ counts from sorter
Tot live cells 3h post sort
Tot sorted cells 4d post sort
B
Pre-sort
50%
Post-sort
83.1%
SSC
mCherry

Fig 5. mCherry sorting to enrich for transduced cells

Intracellular cytokine sorting: sorting the cytokine-positive population (CytX) in two rounds ('yield' followed by 'purity') allows >95% purity (Fig 6).

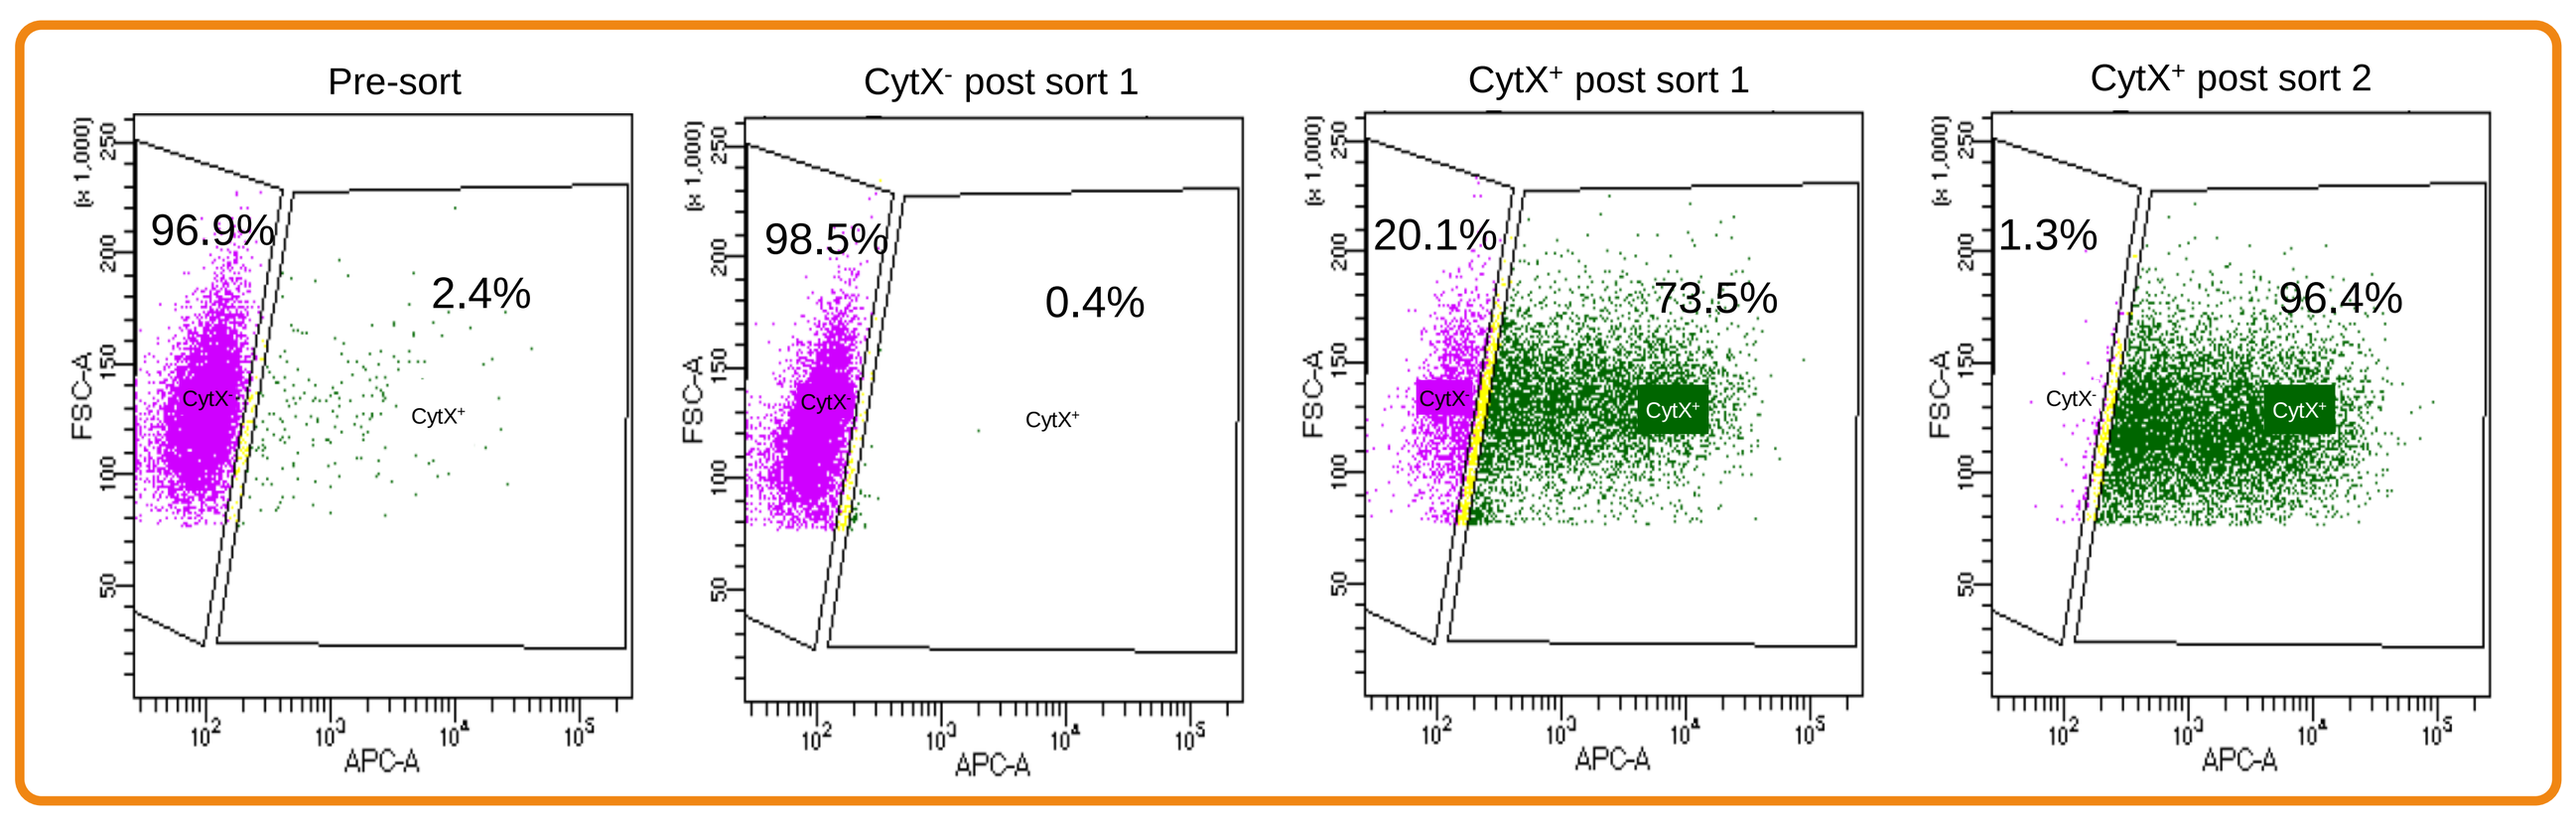
Pre-sort
96.9%
2.4%
CytX- post sort 1
98.5%
0.4%
CytX+ post sort 1
20.1%
73.5%
CytX+ post sort 2
1.3%
96.4%
FSC-A
APC-A

Fig 6. Sorting for an intracellular cytokine

Selection of flow cytometry-based cytokine assays

A flow-based assay was required to detect and sort edited cells producing the anti-inflammatory cytokine. An intracellular and a surface-secreted cytokine staining assay were considered and key differences in their use and outputs are highlighted in Table 1. When staining non-transduced CD4+ cells with both methods, the stain index for the surface-secreted assay was much greater and the staining background (Fig 7) was lower than compared with the intracellular assay. Based on this data and the advantages of the assay, the surface-secreted cytokine assay was selected.

	Intracellular	Surface-secreted
Antibody staining	Requires fix/perm of cells	No fix/perm required
Secretion blocker	May require secretion-blocking compound	No blocker required
Measure of cytokine release	Does ↑ intracellular conc = ↑ release?	Direct measure of release

Table 1. Comparison of an intracellular and a surface-secreted cytokine assay

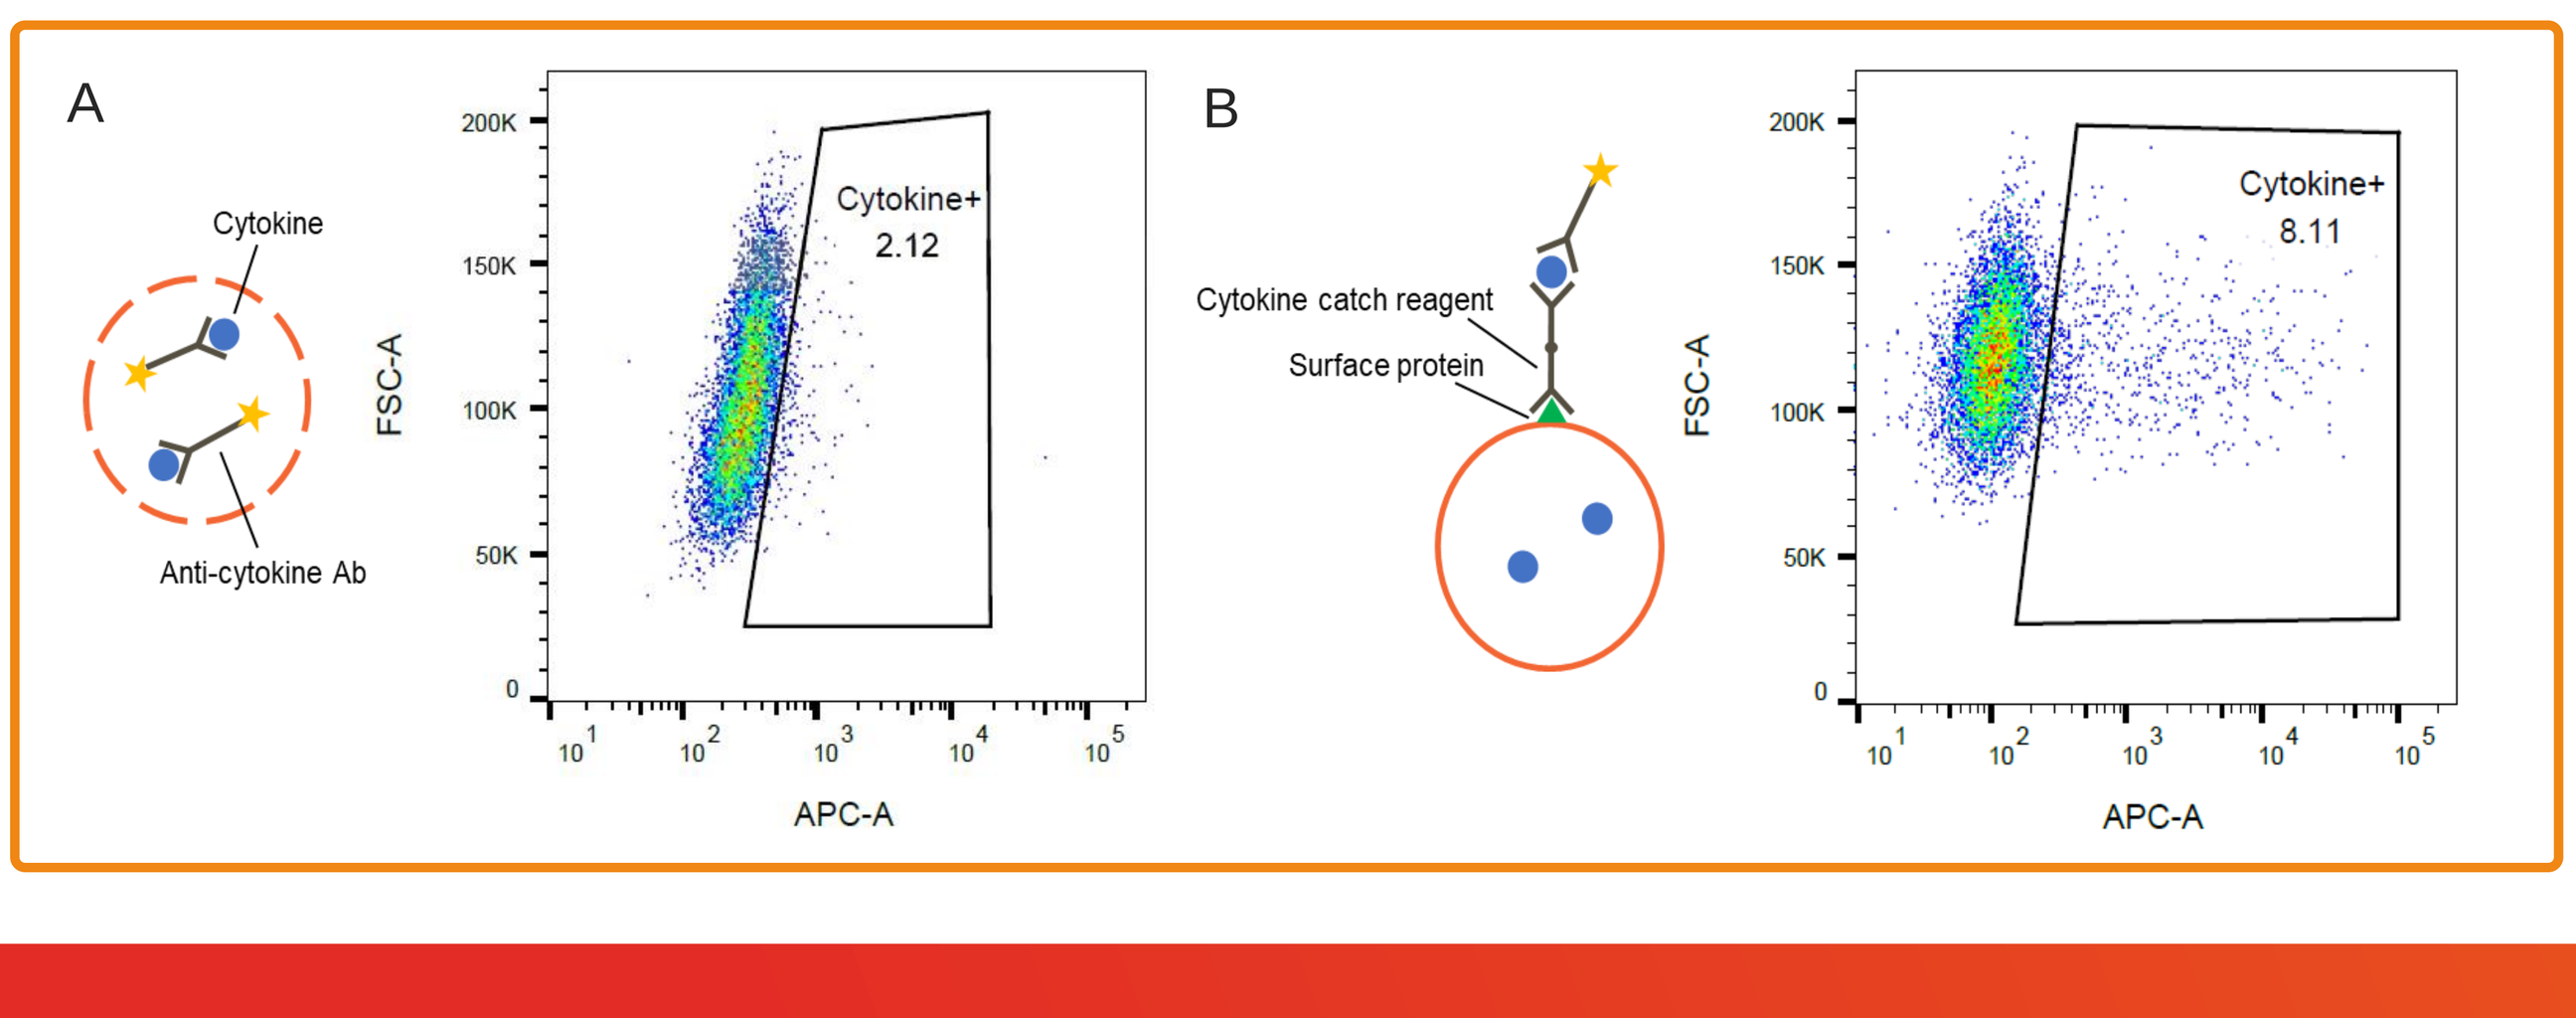
A
Cytokine
Anti-cytokine Ab
Cytokine+
2.12
FSC-A
APC-A
B
Cytokine catch reagent
Surface protein
Cytokine+
8.11
FSC-A
APC-A

Fig 7. Schematics and detection of cytokine-positive cells using the intracellular (A) or surface-secretion (B) cytokine staining assays.

Results II – Pilot Screen

Generating a mini library

To test the optimised CRISPR screening workflow, we ran a pilot screen in T cells from two healthy donors using a "mini library" comprising 1,000 gRNAs, a fifth of which are known to lead to upregulation of the cytokine (literature evidence and previous GSK screening efforts) (Table 2). Fig 8 outlines how the mini library was generated.

	Genes	gRNAs
↑ cytokine*	36	208
↓ cytokine*	10	60
no effect on cytokine*	80	462
non-targeting	N/A	200
introns	9	72
TOTAL	126	1002

*determined from a previous RNP-based arrayed CRISPR screen (GSK)

Table 2. Mini library composition

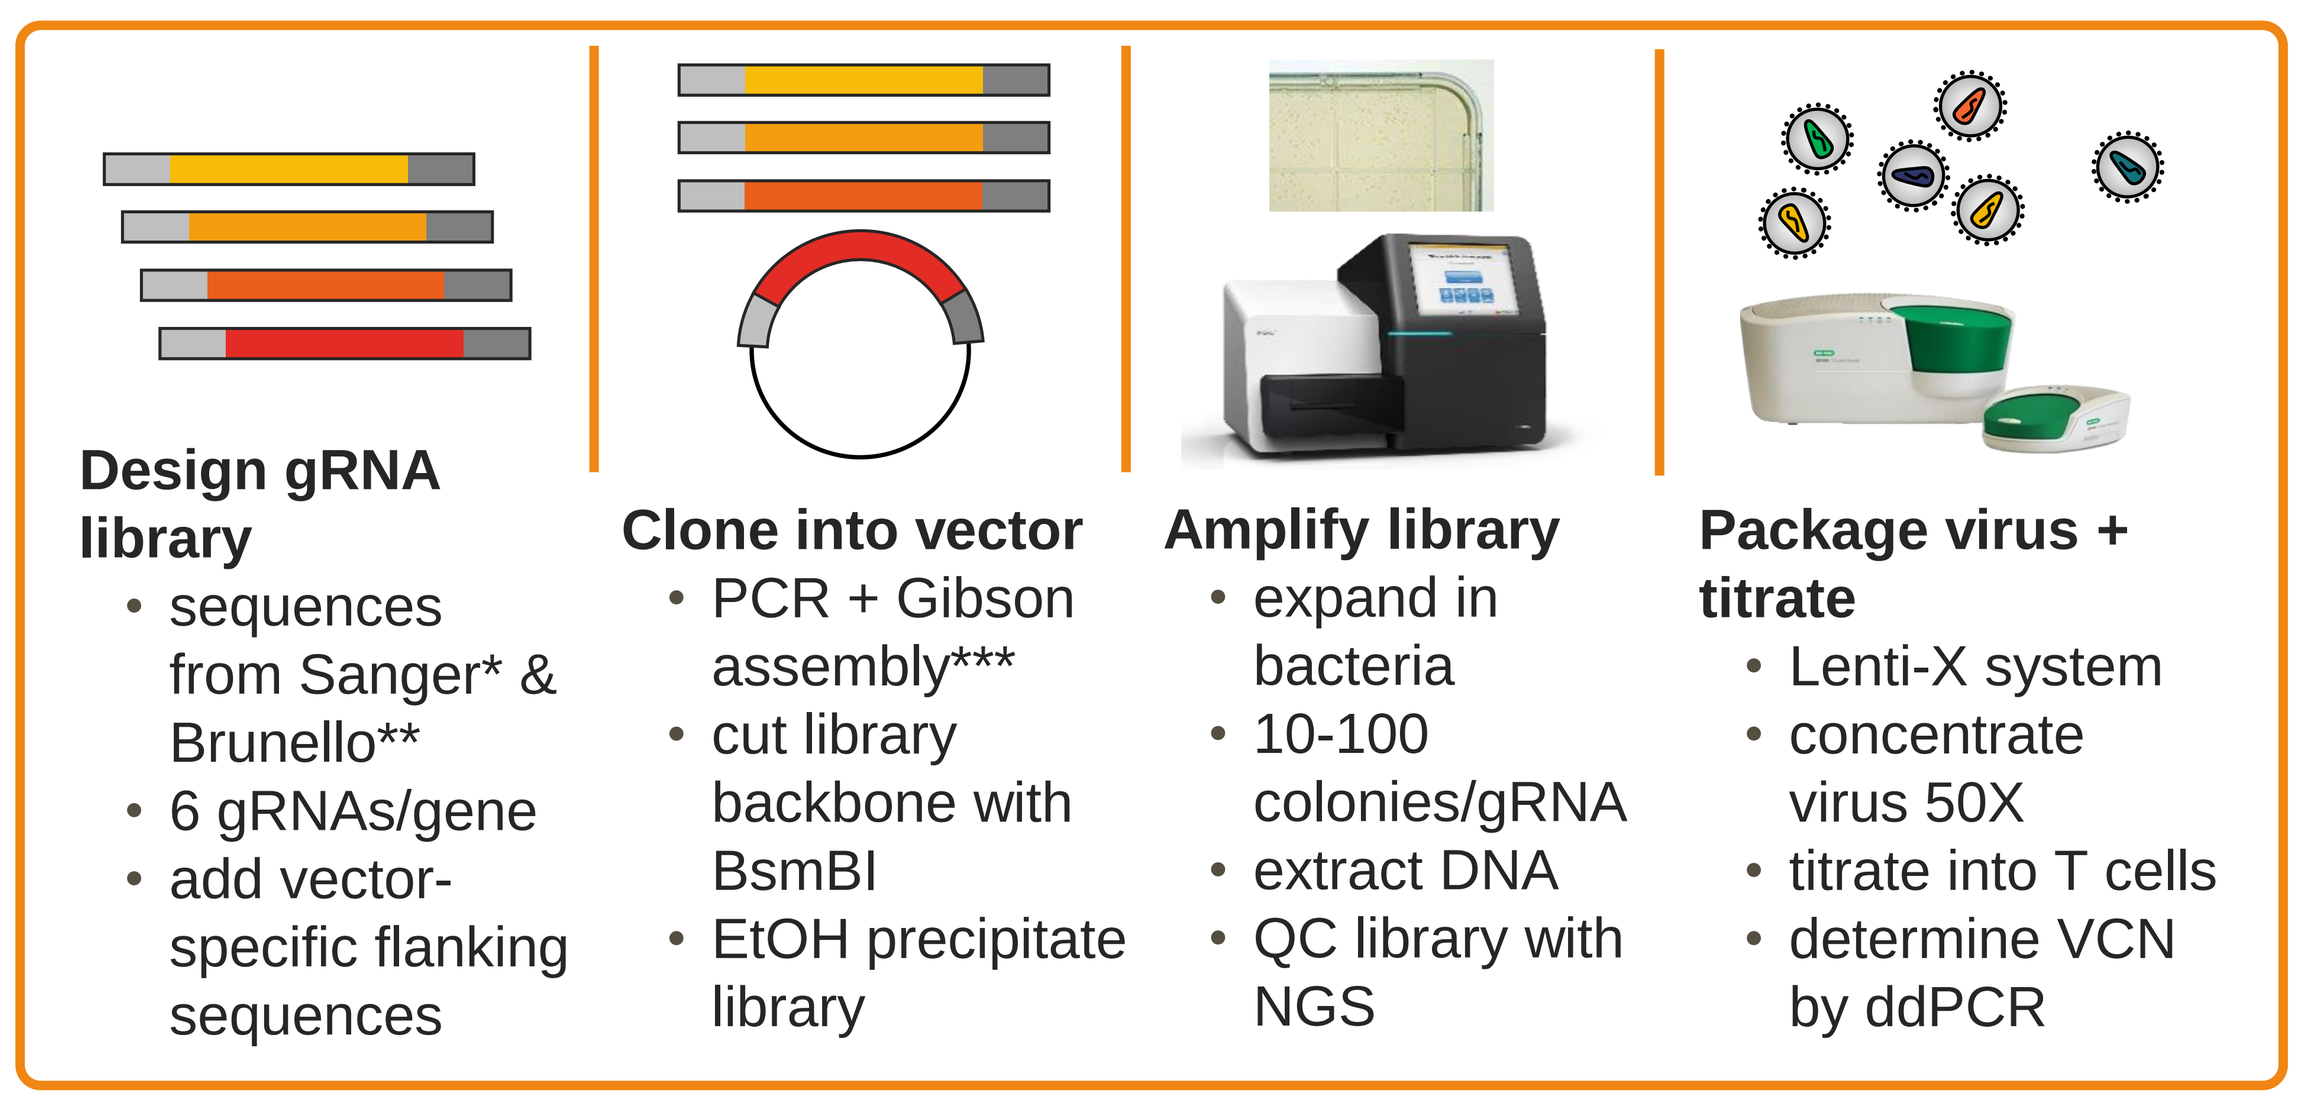
Design gRNA library
sequences from Sanger* & Brunello**
6 gRNAs/gene
add vector-specific flanking sequences
Clone into vector
PCR + Gibson assembly***
cut library backbone with BsmBI
EtOH precipitate library
Amplify library
expand in bacteria
10-100 colonies/gRNA
extract DNA
QC library with NGS
Package virus + titrate
Lenti-X system
concentrate virus 50X
titrate into T cells
determine VCN by ddPCR

Fig 8. Generation of mini library

*Genome Biol. 2021 Jan 21;22(1):40
**Nat. Commun. 2018 Dec 21;9(1):5416
***Nat Protoc. 2017 Apr;12(4):828-863

ddPCR data showed that a ~20% transduction efficiency was required to achieve ~1 gRNA/cell. We aimed to maintain >400X representation of the library at all times throughout the screen. Fig 9 shows the experimental design of the focused screen.

Running the screen

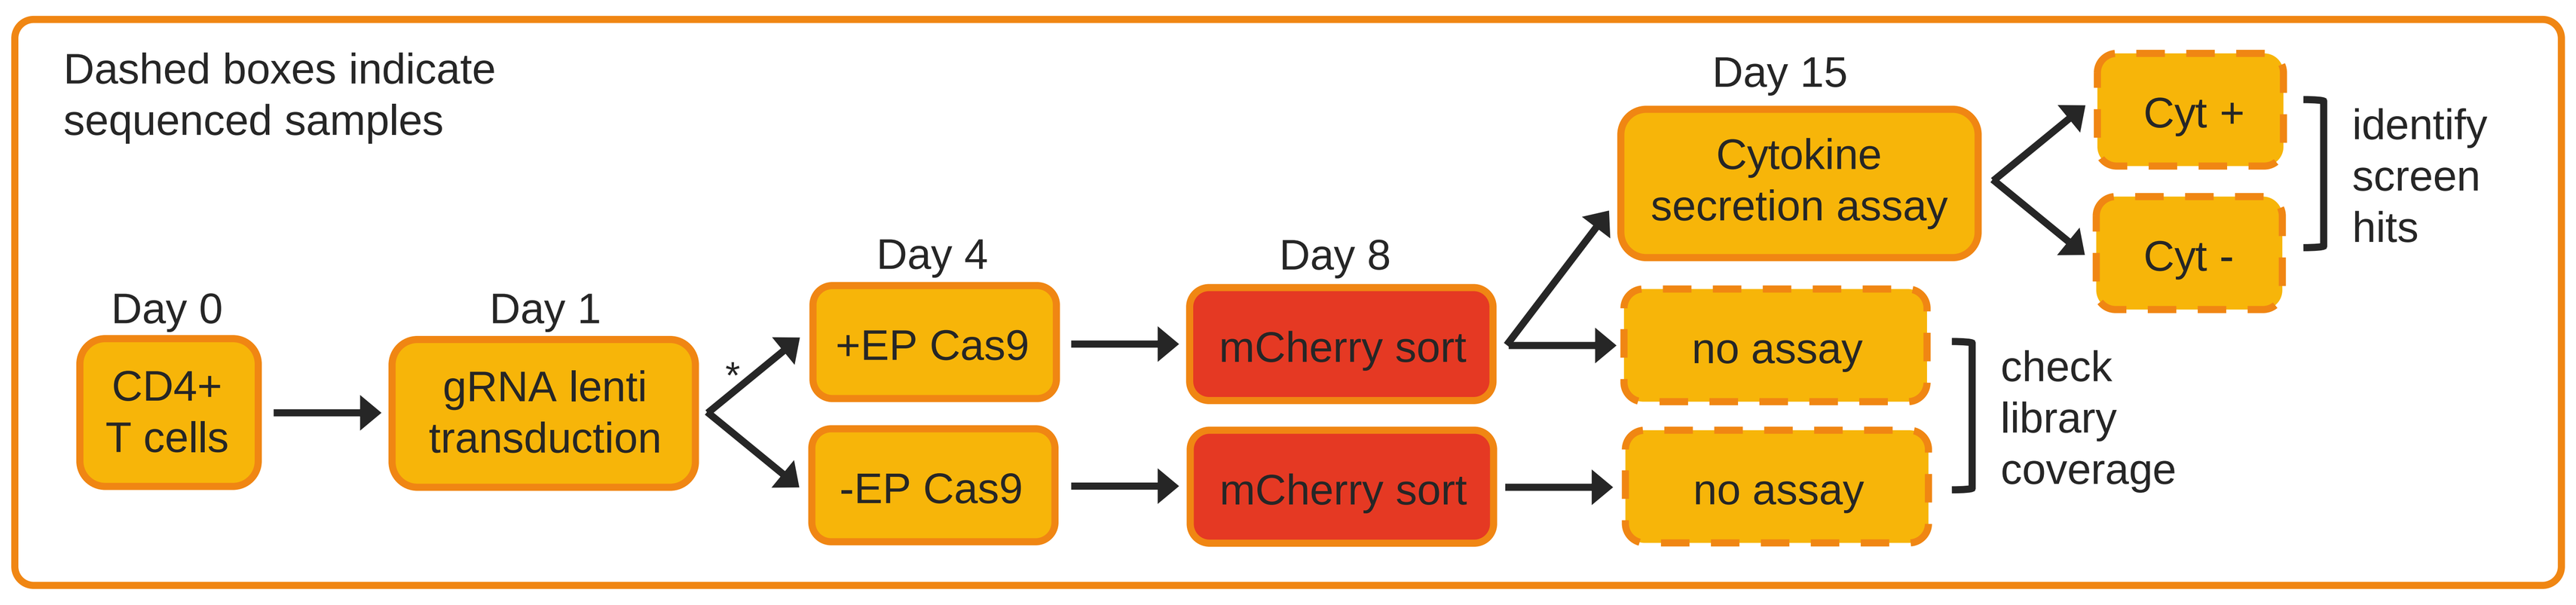
Dashed boxes indicate sequenced samples
Day 0
CD4+ T cells
Day 1
gRNA lenti transduction
Day 4
+EP Cas9
-EP Cas9
Day 8
mCherry sort
mCherry sort
Day 15
Cytokine secretion assay
Cyt +
Cyt -
identify screen hits
no assay
no assay
check library coverage

Fig 9. Experimental design of mini screens

A QC edit of B2M 4 days post-EP showed ~80% knockout, suggesting Cas9 electroporation was efficient in SLICE cells. On day 8, we achieved >3-fold enrichment of mCherry+ cells by FACS. On day 15, the secretion assay showed that 7.8% of cells were positive for the cytokine. We achieved 96.4% purity of cytokine-positive cells after FACS.

NGS libraries were prepared and sequenced, and raw data passed key QC analyses (% mapped reads, Gini index and library coverage). Log fold-change analysis of raw gRNA counts showed the expected changes in gRNA representation (Fig 10A) and we observed good reproducibility in guide counts between the donors (Fig 10B). Counts were processed with LimmaVoom, an R: based tool used to determine the fold change and significance of enrichment of each gRNA between cytokine-positive and negative populations (Fig 10C). Encouragingly, the top 4 ranked genes were strong positive hits from the RNP-based arrayed CRISPR screen (internal GSK data).

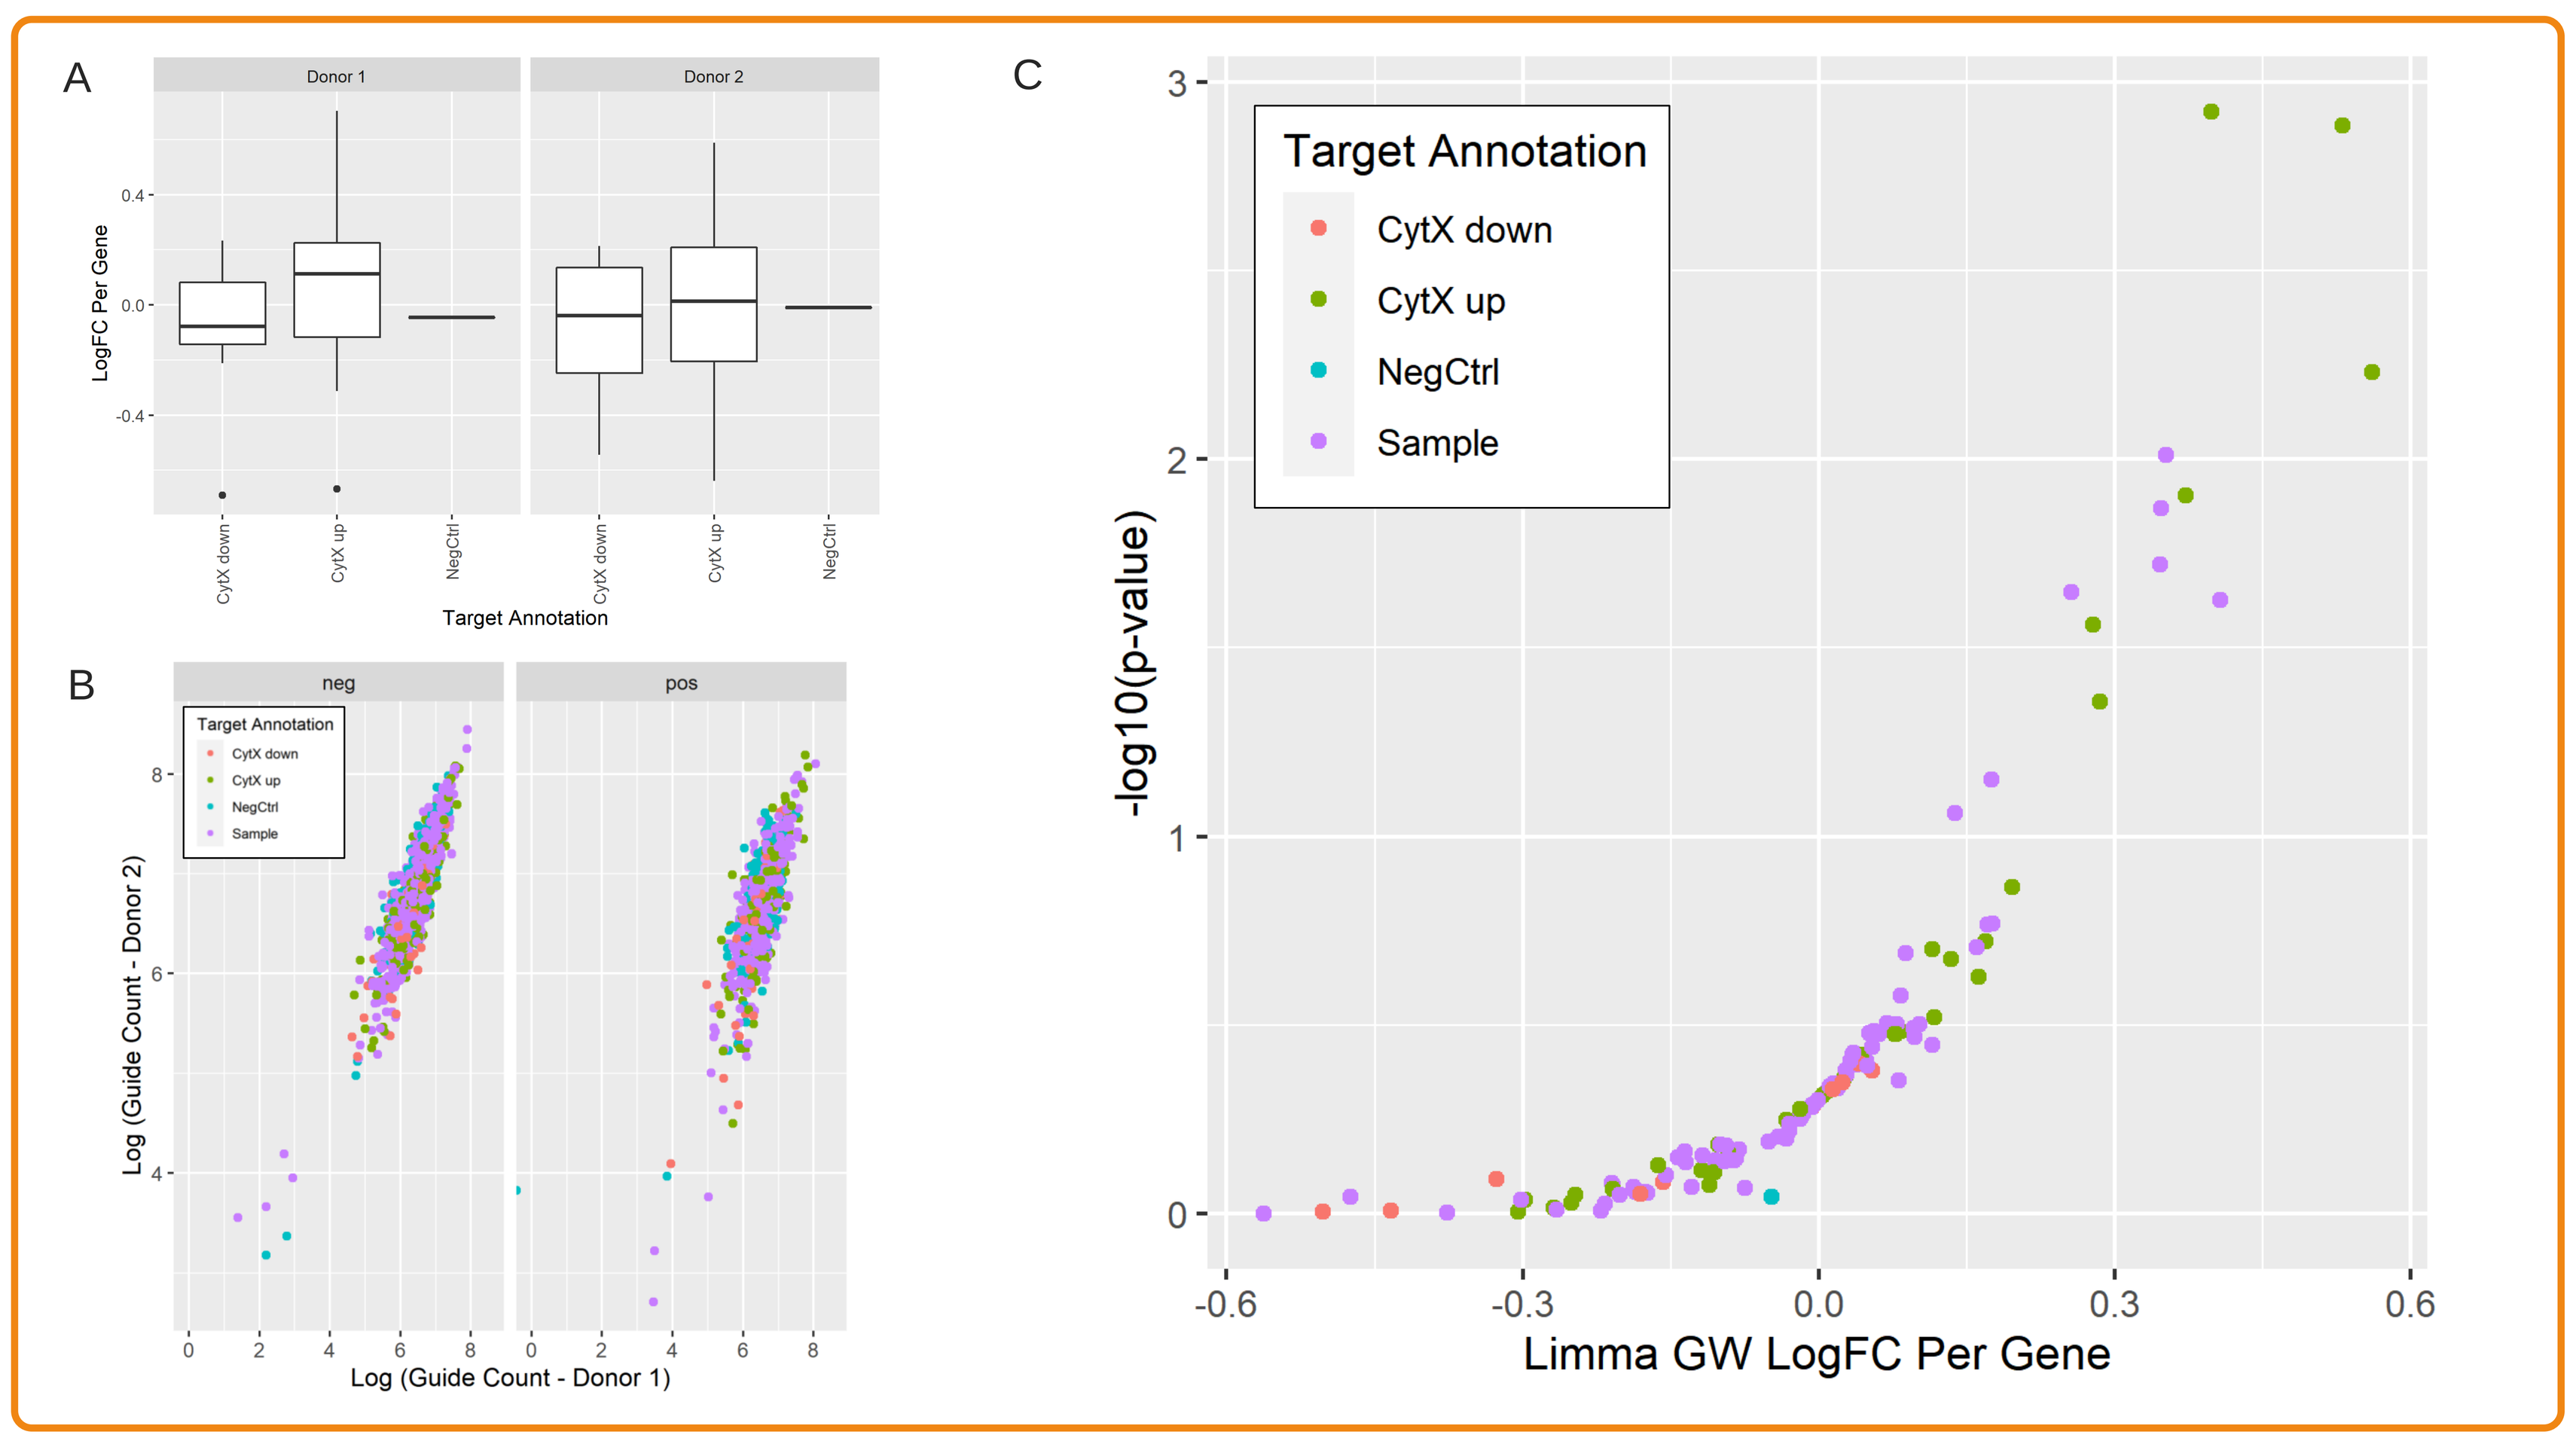
A
Donor 1
Donor 2
LogFC Per Gene
Target Annotation
B
neg
pos
Log (Guide Count - Donor 2)
Log (Guide Count - Donor 1)
C
Target Annotation
CytX down
CytX up
NegCtrl
Sample
-log10(p-value)
Limma GW LogFC Per Gene

Fig 10. LogFC gRNA plots (A), inter-donor guide count correlation (B) and LimmaVoom gRNA enrichment analysis (C)

Conclusions

- We successfully optimised key elements of the pooled CRISPR workflow and compared the performance of FACS assays for both intracellular and surface-secreted cytokine
- We produced a focused gRNA library and used it to demonstrate good performance of the pooled CRISPR workflow, observing an enrichment of cytokine-increasing guides in the cytokine-positive cell population
- We will now execute a genome-wide screen in three CD4+ T cell donors