CATAPULT Medicines Discovery

Investigation of Nuvec Particles for Effective Gene Delivery

N. Ponnappan[1], J. Szczerkowski[1], M. Bachman[1], M. Main[1], D. Templeton[2], R. Harris[2], M. Janssen[2], N. Theobald[2], D. Yilmaz[1]

1. Medicines Discovery Catapult, Alderley Park, Cheshire, SK10 4ZF, UK.

2. N4 Pharma, Weston house Bradgate Park View, Chellaston, Derbyshire, DE73 5UJ

md.catapult.org.uk

Abstract

Effective delivery of nucleic acids into the cells holds great potential for vaccine development and cancer treatments. N4 Pharma has been developing Nuvec® – a unique non-viral adjuvant delivery system for the effective delivery of nucleic acids into the cells. Nuvec silica nanoparticles have a unique irregular (spiky) surface structure, coupled with polyethyleneimine (PEI), that simply and effectively traps and protects nucleic acids (such as mRNA / pDNA) as it travels into the cells.

Here we carried out biological characterisation of Nuvec particles using a range of methods. We determined the size, zeta potential and DNA/RNA loading capacity of different size Nuvec particles using DLS, zeta sizer and mass spectrometry respectively. Next, we determined the transfection efficiency of Nuvec at different Nuvec:nucleic acid ratios using both live cell imaging and flow cytometry methods. Furthermore, we investigated the stability of mRNA upon lyophilisation of Nuvec:mRNA complex and demonstrated that Nuvec particles protect the RNA from degradation even after lyophilisation. Additionally, we studied the cellular internalisation of Nuvec using high resolution microscopy. We analysed 3D reconstructed microscopy images and quantified the number of Nuvec particles inside the cells. Finally, we studied the mechanism of cellular internalisation in HEK293T cells upon inhibition of different endocytosis pathways.

Our data demonstrates that Nuvec particles provide protection to RNA/DNA, efficiently deliver DNA/RNA into the cells and result in protein expression. Furthermore, our data provides insights into the mechanisms involved in the cellular uptake of Nuvec particles. Altogether our findings provide guidance for the rational design of Nuvec for efficient gene delivery applications.

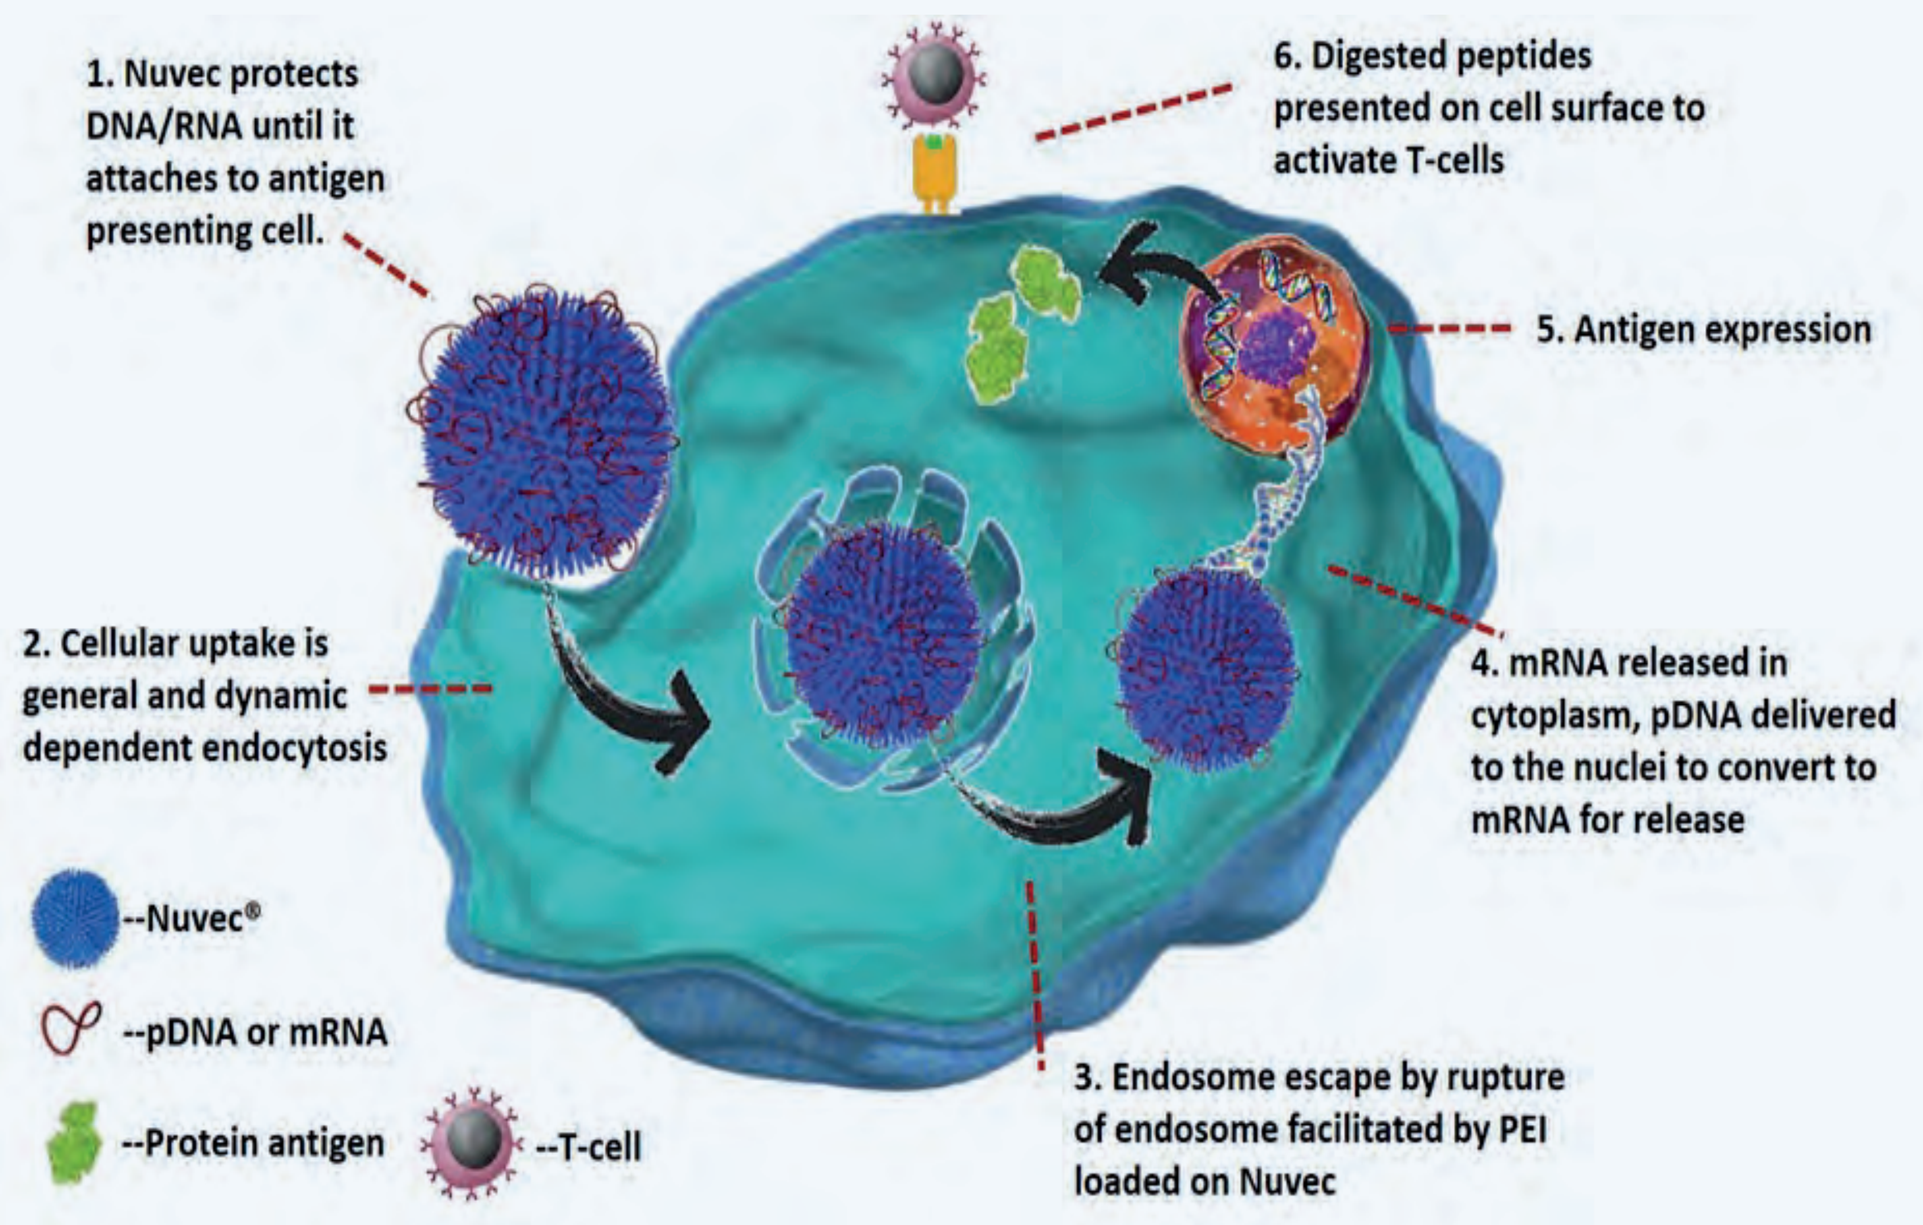

1. Nuvec protects DNA/RNA until it attaches to antigen presenting cell.
2. Cellular uptake is general and dynamic dependent endocytosis
3. Endosome escape by rupture of endosome facilitated by PEI loaded on Nuvec
4. mRNA released in cytoplasm, pDNA delivered to the nuclei to convert to mRNA for release
5. Antigen expression
6. Digested peptides presented on cell surface to activate T-cells
--Nuvec®
--pDNA or mRNA
--Protein antigen
--T-cell

Method

- Loading of Nuvec with different amounts of plasmid DNA/mRNA
- Incubation at room temperature for 20 min
- Lyophilisation of complex
- Size distribution and zeta potential analysis using DLS & zetasizer
- Mass spectrometry analysis to assess the binding of DNA/mRNA molecule to Nuvec particles
- Transfection of HEK293T cells seeded in 96-well plate with nucleic acid: Nuvec complex
- Incubation at 37°C, 5% $C0_2$ for different time-periods
- Microscopy and flow cytometry analysis

Results

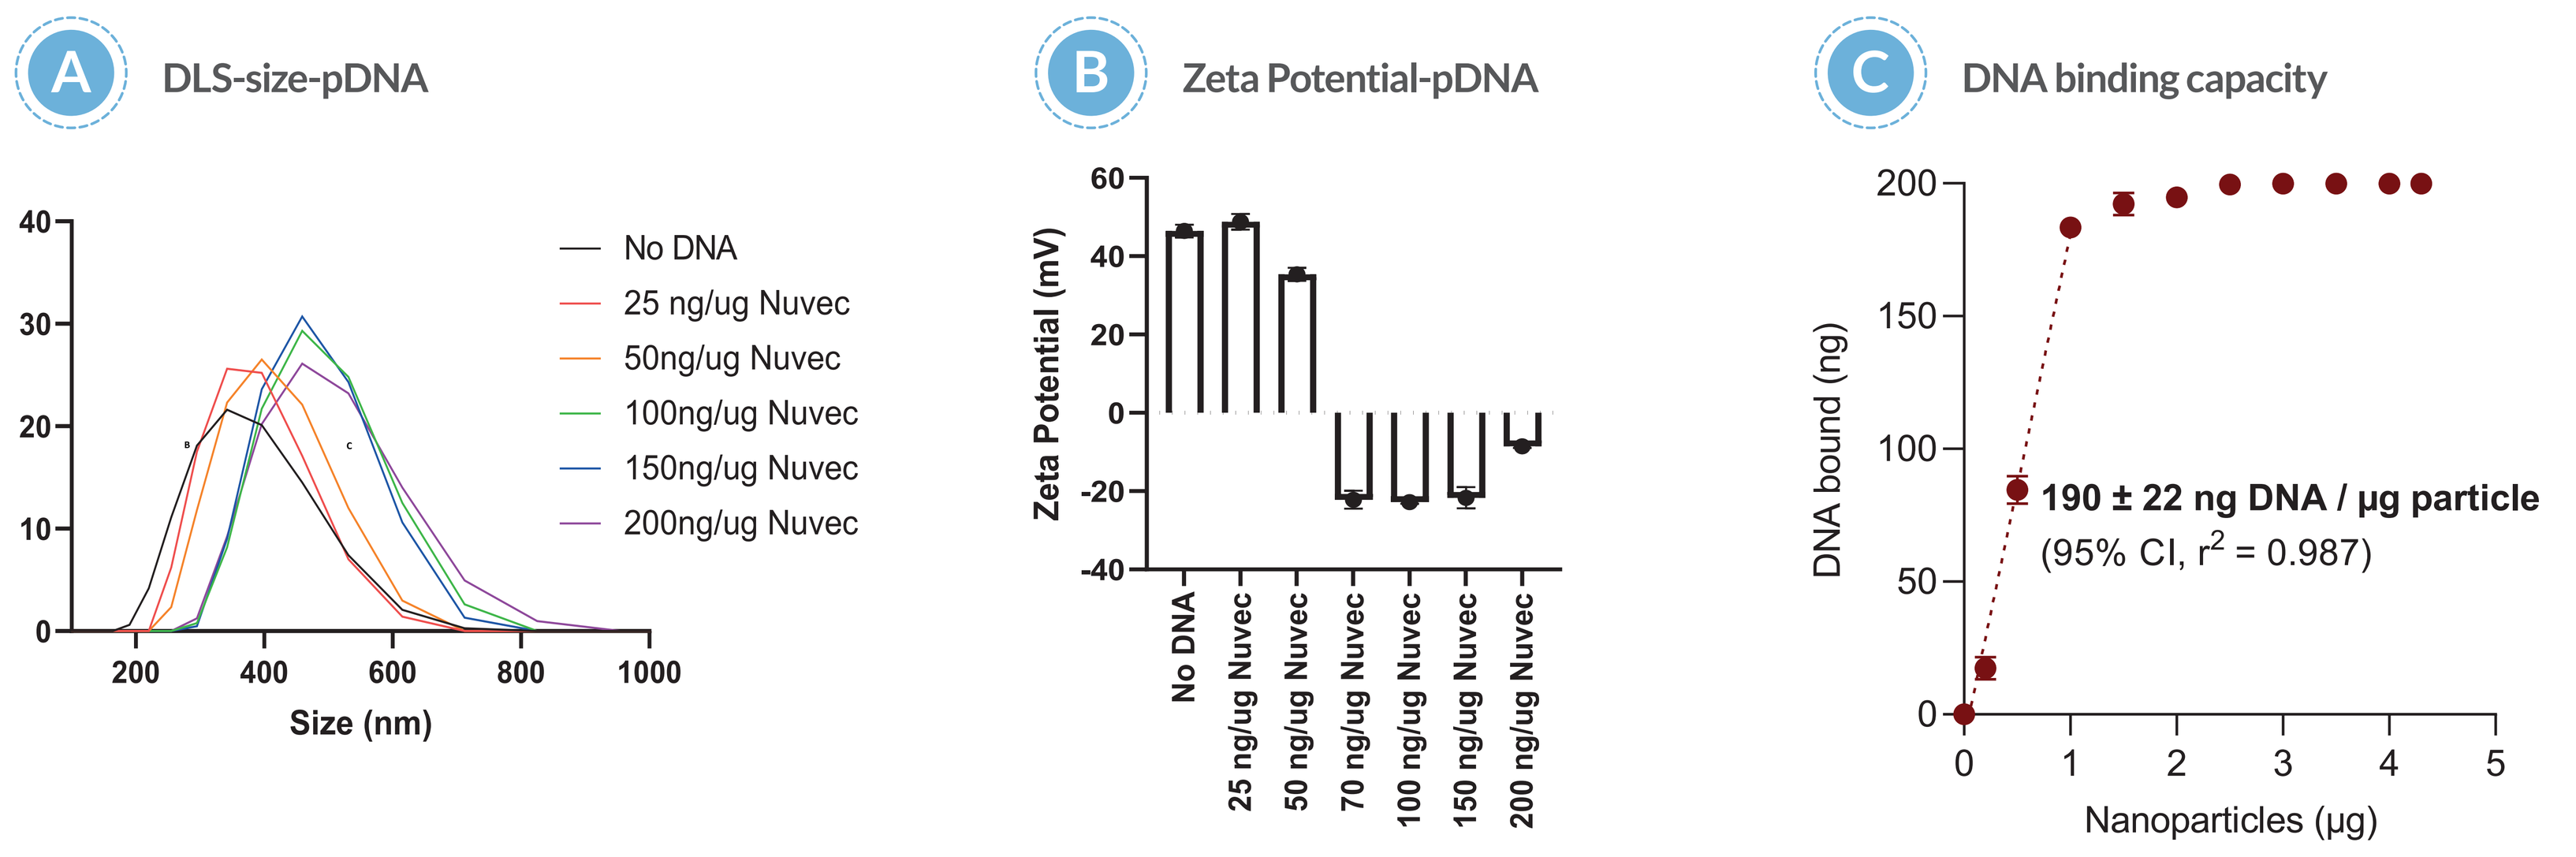

A
DLS-size-pDNA
No DNA
25 ng/ug Nuvec
50ng/ug Nuvec
100ng/ug Nuvec
150ng/ug Nuvec
200ng/ug Nuvec
Size (nm)
B
Zeta Potential-pDNA
Zeta Potential (mV)
No DNA
25 ng/ug Nuvec
50 ng/ug Nuvec
70 ng/ug Nuvec
100 ng/ug Nuvec
150 ng/ug Nuvec
200 ng/ug Nuvec
C
DNA binding capacity
DNA bound (ng)
190 ± 22 ng DNA / µg particle
(95% CI, r2 = 0.987)
Nanoparticles (µg)

Figure 1. Plasmid DNA (pmaxGFP) loading on Nuvec particles at various mass ratios (ng of plasmid per ug of Nuvec). A: Size distribution analysis of pmaxGFP:Nuvec particles by DLS. B: Assessment of zeta potential of particles by zeta nanosizer. C: Mass spectrometry analysis of binding of plasmid DNA to Nuvec particles.

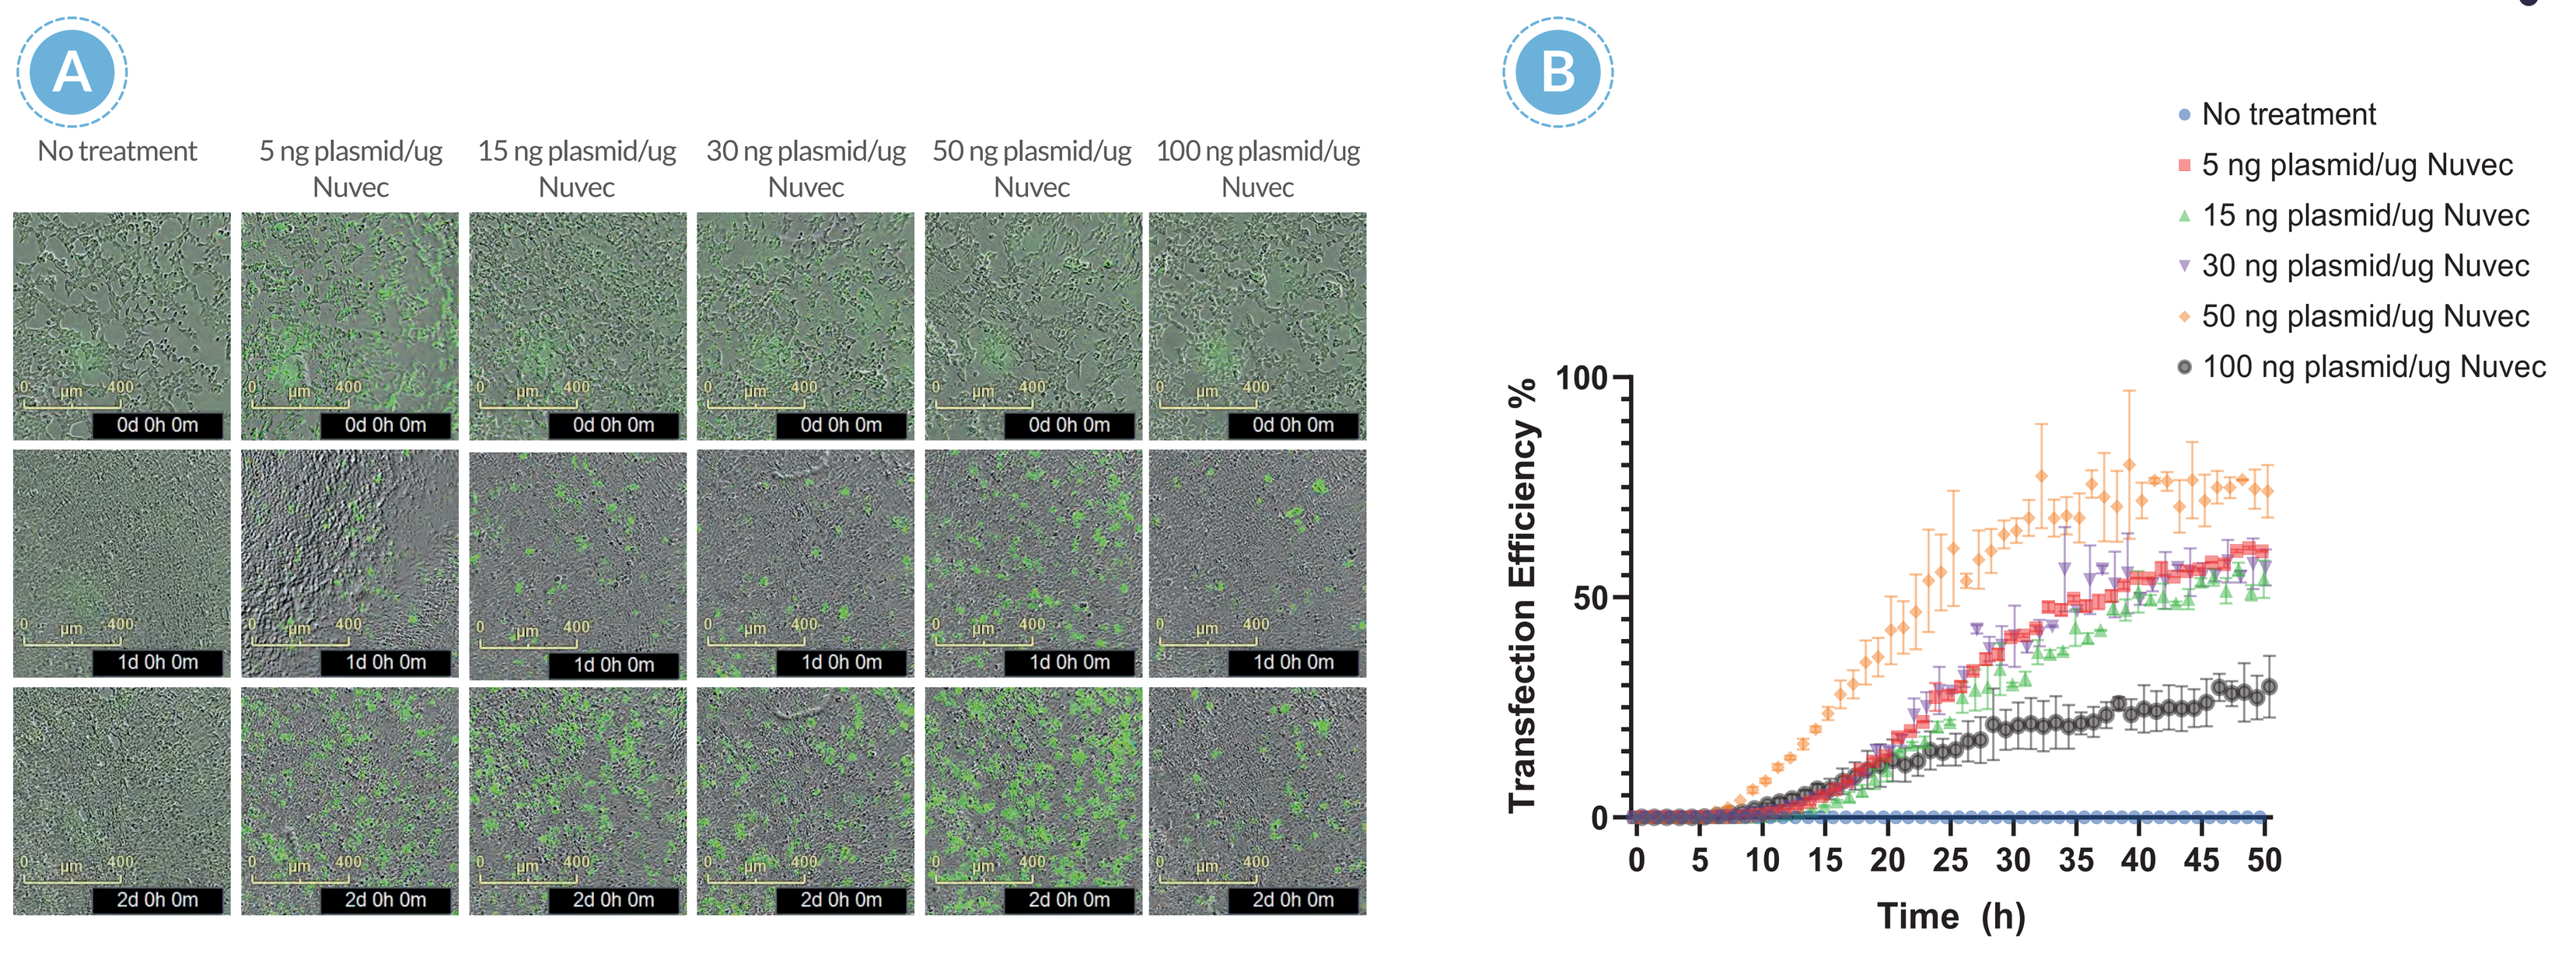

A
No treatment
5 ng plasmid/ug Nuvec
15 ng plasmid/ug Nuvec
30 ng plasmid/ug Nuvec
50 ng plasmid/ug Nuvec
100 ng plasmid/ug Nuvec
0d 0h 0m
1d 0h 0m
2d 0h 0m
B
No treatment
5 ng plasmid/ug Nuvec
15 ng plasmid/ug Nuvec
30 ng plasmid/ug Nuvec
50 ng plasmid/ug Nuvec
100 ng plasmid/ug Nuvec
Transfection Efficiency %
Time (h)

Figure 2. Representative microscopy images (A) & graph showing the transfection efficiency (B) of HEK293T cells transfected with plasmid:Nuvec at different mass ratios over a period of 48 h.

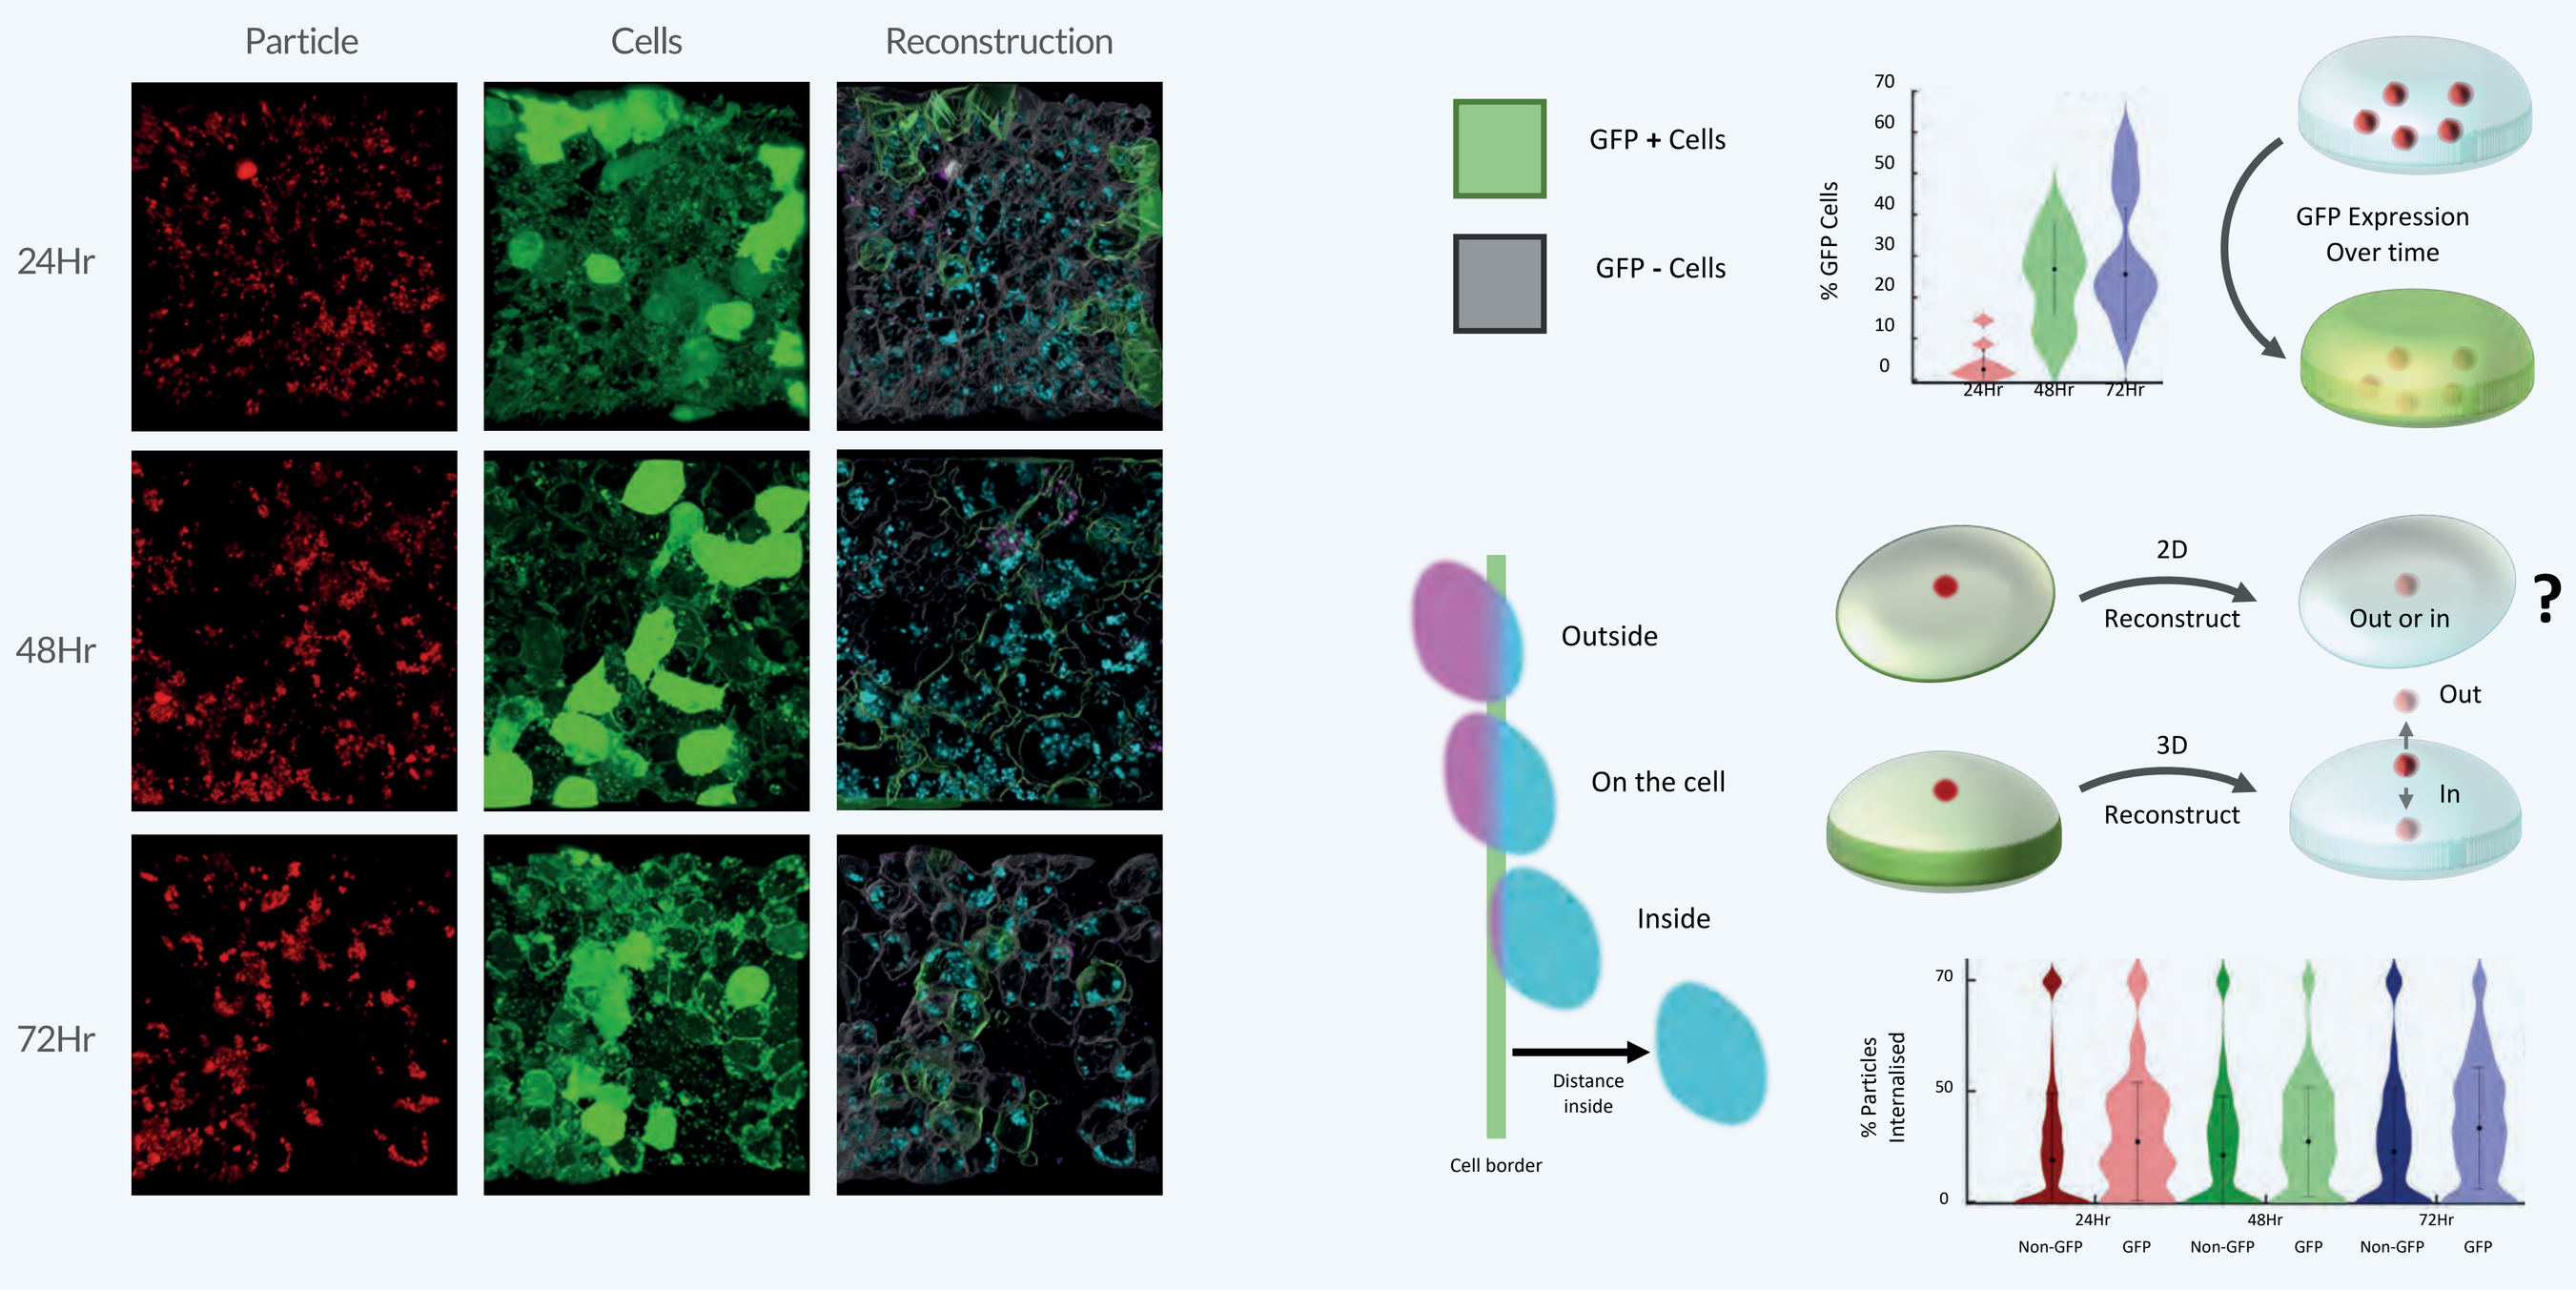

Particle
Cells
Reconstruction
24Hr
48Hr
72Hr
GFP + Cells
GFP - Cells
% GFP Cells
GFP Expression Over time
Outside
On the cell
Inside
Distance inside
Cell border
2D
Reconstruct
Out or in
?
3D
Reconstruct
Out
In
% Particles internalised
Non-GFP
GFP

Figure 3. High resolution microscopy images of HEK293T cells treated with Alexa-568 labelled Nuvec particles. 3D reconstruction of images were carried out and percentage of cells expressing GFP and number of Nuvec particles internalised were assessed

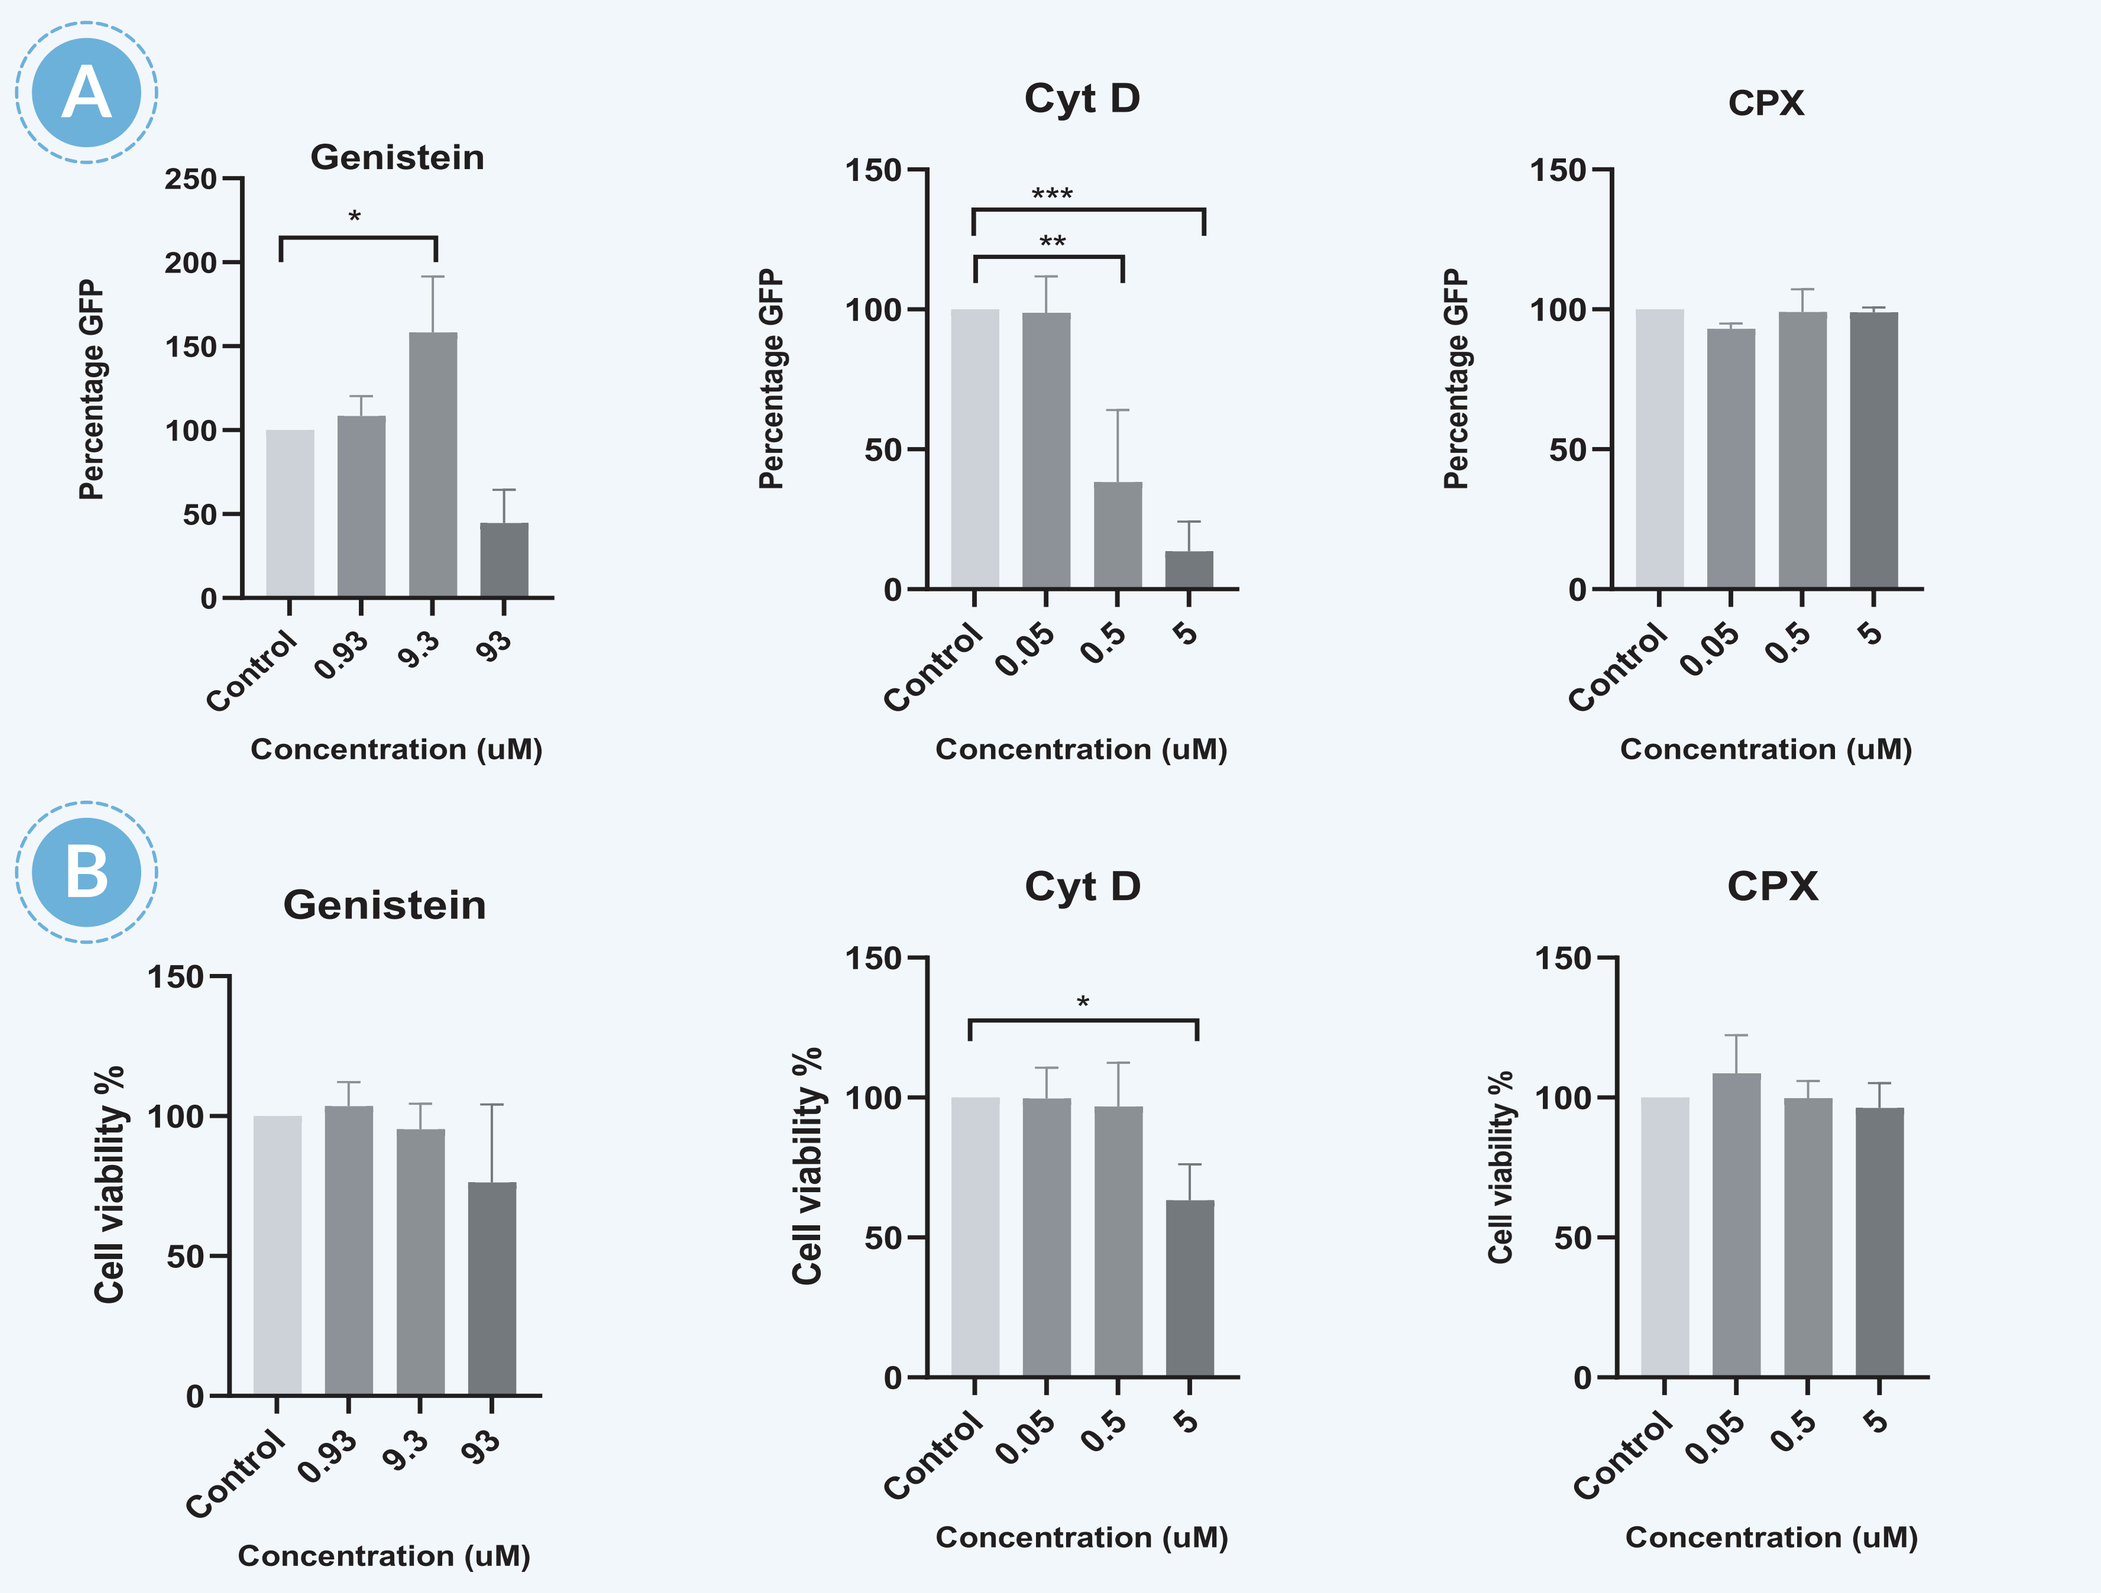

A
Genistein
Percentage GFP
Control
0.93
9.3
93
Concentration (uM)
Cyt D
0.05
0.5
5
CPX
B
Cell viability %

Figure 4. Effect of endocytic inhibitors on the transfection efficiency of plasmid DNA loaded Nuvec (A) & viability of cells (B). Control are the HEK293T cells with no endocytic inhibitor.

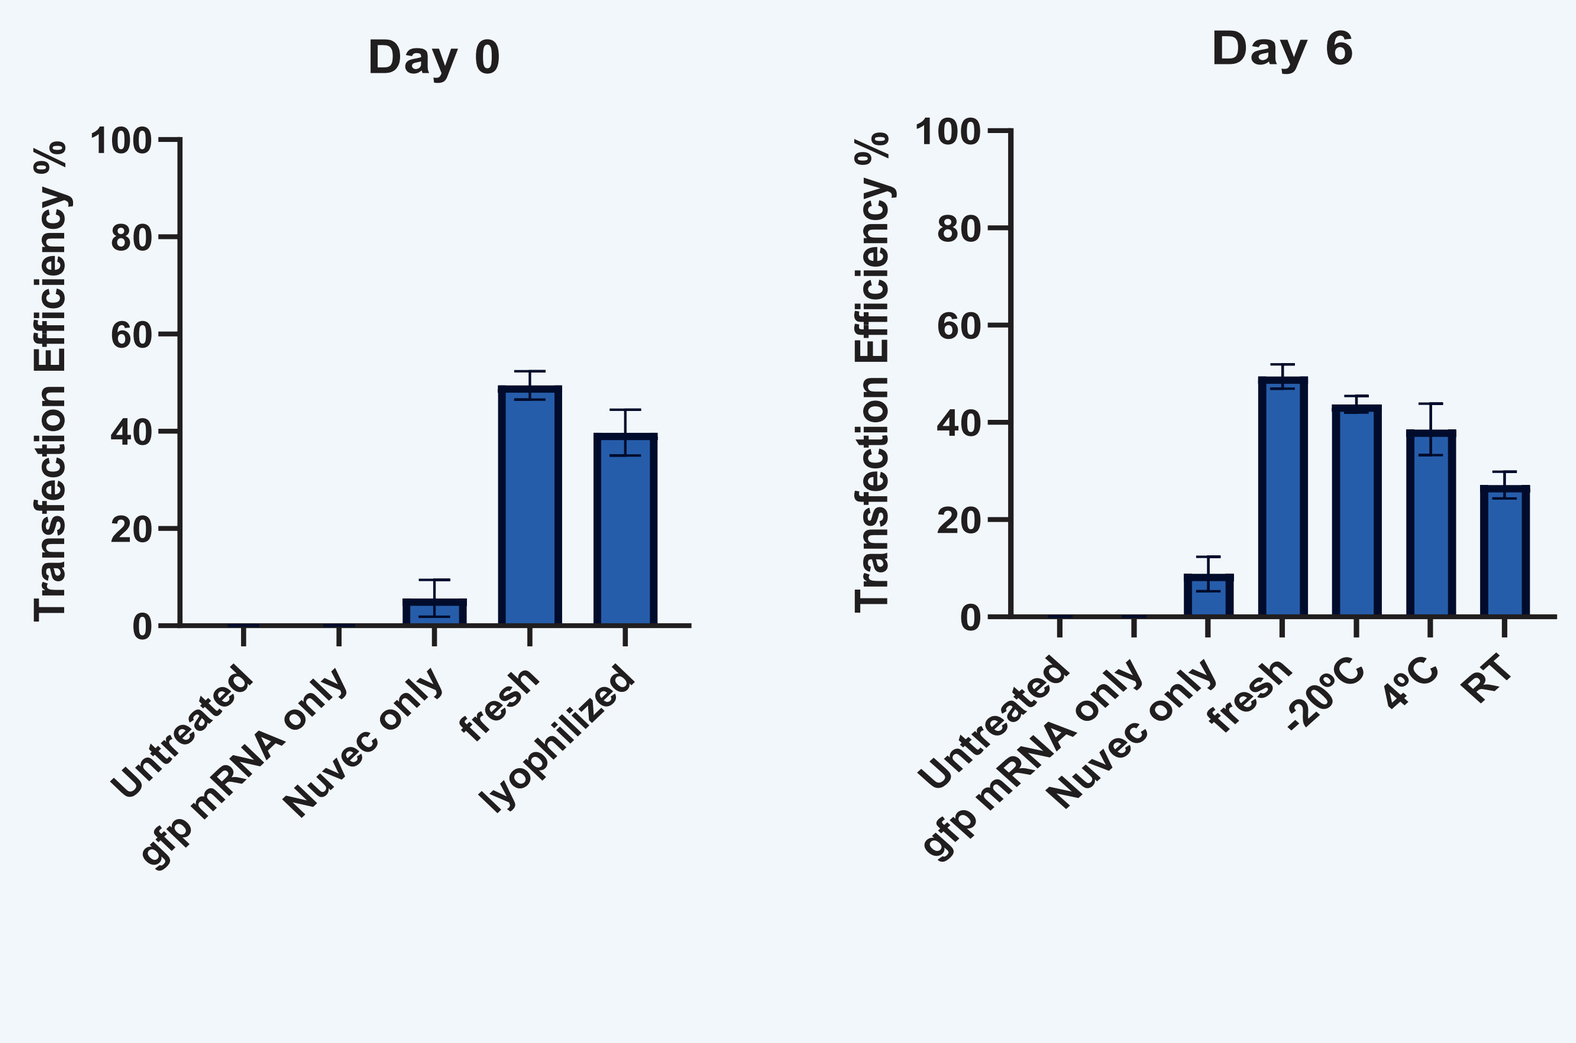

Day 0
Day 6
Transfection Efficiency %
Untreated
gfp mRNA only
Nuvec only
fresh
lyophilized
-20°C
4°C
RT

Figure 5. Transfection efficiency of freshly lyophilized GFP mRNA:Nuvec at day 0 and that stored at different conditions for 6 days after lyophilization

Conclusions

- Nuvec particle efficiently binds to nucleic acids – plasmid DNA & mRNA, and mediates their intracellular delivery mainly through macropinocytic pathway
- The cells transfected with GFP plasmid loaded Nuvec particles exhibit up to 70% transfection efficiency
- Our data for transfection efficiency of lyophilized GFP mRNA loaded Nuvec stored at -20°C indicates the ability of Nuvec particles to protect the nucleic acids from degradation. The freeze-dried formulations of Nuvec-based nucleic acid therapeutics would be convenient and cost-effective

Medicines Discovery Catapult (MDC)

Funded by Innovate UK, MDC is national centre set up to help UK SMEs, biotechs, academics and innovators with access to cutting edge technology, expertise, data and networks they need to progress their drug discovery programs.

Through collaborative programmes of R&D we are tackling the most challenging issues in drug discovery, addressing systemic problems and bottlenecks and using innovative technologies to enable “fast-to-patient” medicines discovery.

Innovate UK