Development of a kinetic ATPase assay for mechanism of inhibition studies

Chris Pedder[1], Claire McWhirter[2], Amanda Ferdinand[1], Tim Hammonds[1]

[1]Cancer Research UK Therapeutic Discovery Laboratories, Building 910, Babraham Research Campus, Cambridge, CB22 3AT, United Kingdom

[2]Artios Pharma Limited, Babraham Research Campus, Cambridge, CB22 3AT, United Kingdom

INTRODUCTION

ATPases are involved in multiple aspects of cellular function and are attractive targets for novel cancer therapeutics. Key to designing new drugs targeting ATPases is the ability to measure inhibition of ATP turnover. We routinely use the endpoint ADP Glo assay for ATPase activity that is well suited to screening, however a kinetic assay would be better suited to complex mechanism of inhibition (MoI) studies.

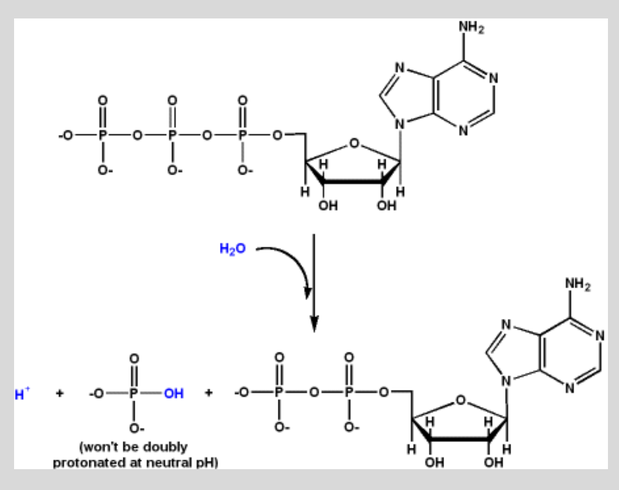

Figure 1. ATP hydrolysis schematic. ATPases catalyse the hydrolysis of adenosine triphosphate (ATP) to adenosine diphosphate (ADP) and inorganic phosphate (P_i). The energy from ATP hydrolysis is utilised by the enzyme to achieve a cellular function.

INITIAL ASSAY DEVELOPMENT

Initially four kinetic ATPase assay formats were investigated; three Transcreener assays from Bellbrooks to measure ADP levels and the phosphate sensor reagent from Thermo Fisher to measure P_i levels.

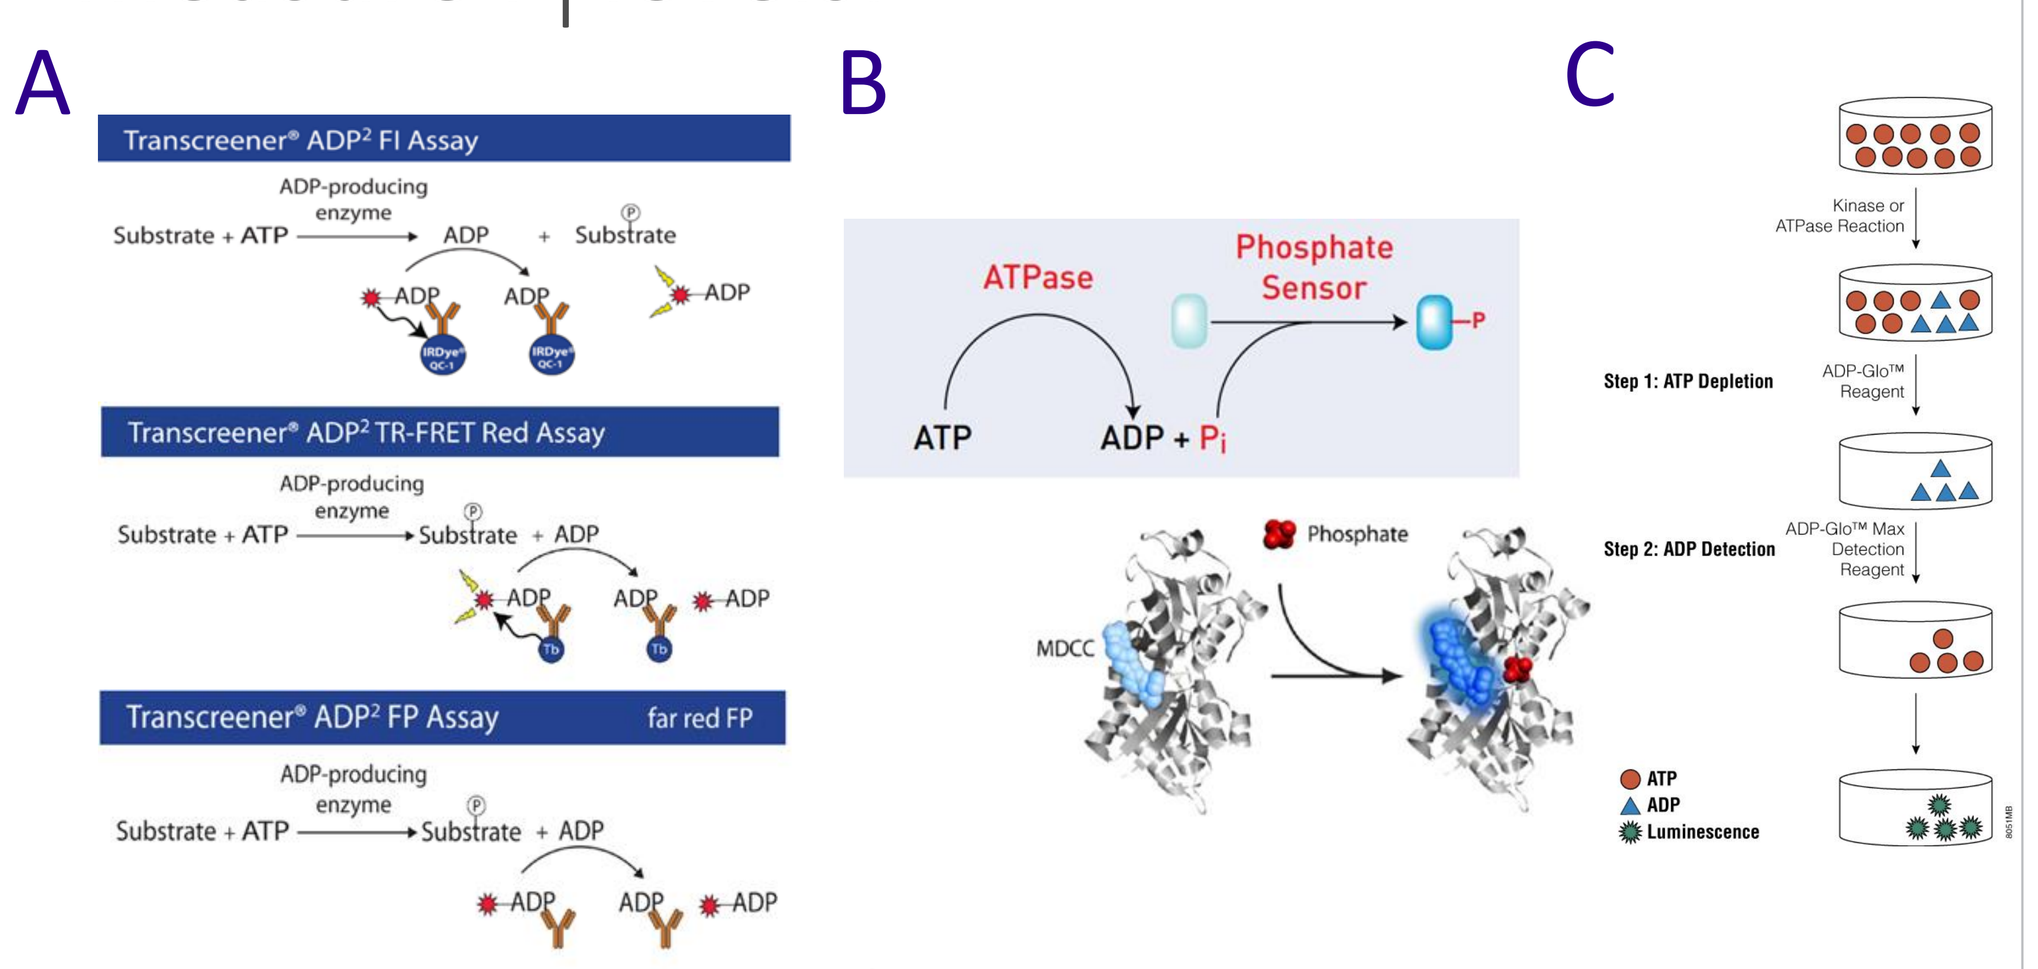

A
B
C
Transcreener® ADP² FI Assay
Transcreener® ADP² TR-FRET Red Assay
Transcreener® ADP² FP Assay
far red FP
ATPase
Phosphate Sensor
ATP
ADP + Pi
MDCC
Phosphate
Step 1: ATP Depletion
Step 2: ADP Detection
ATP
ADP
Luminescence

Figure 2. Schematics for A) the three Bellbrooks Transcreener kinetic ADP detection assays, B) the Thermo Fisher phosphate sensor reagent and C) the endpoint ADP Glo assay.

As part of the initial assay development a compound interference check was performed in each assay format. 21%, 14% and 36% (FI, TR-FRET, FP respectively) of the 14 compounds tested showed compound interference in the Transcreener assays compared to 0% in the phosphate sensor assay.

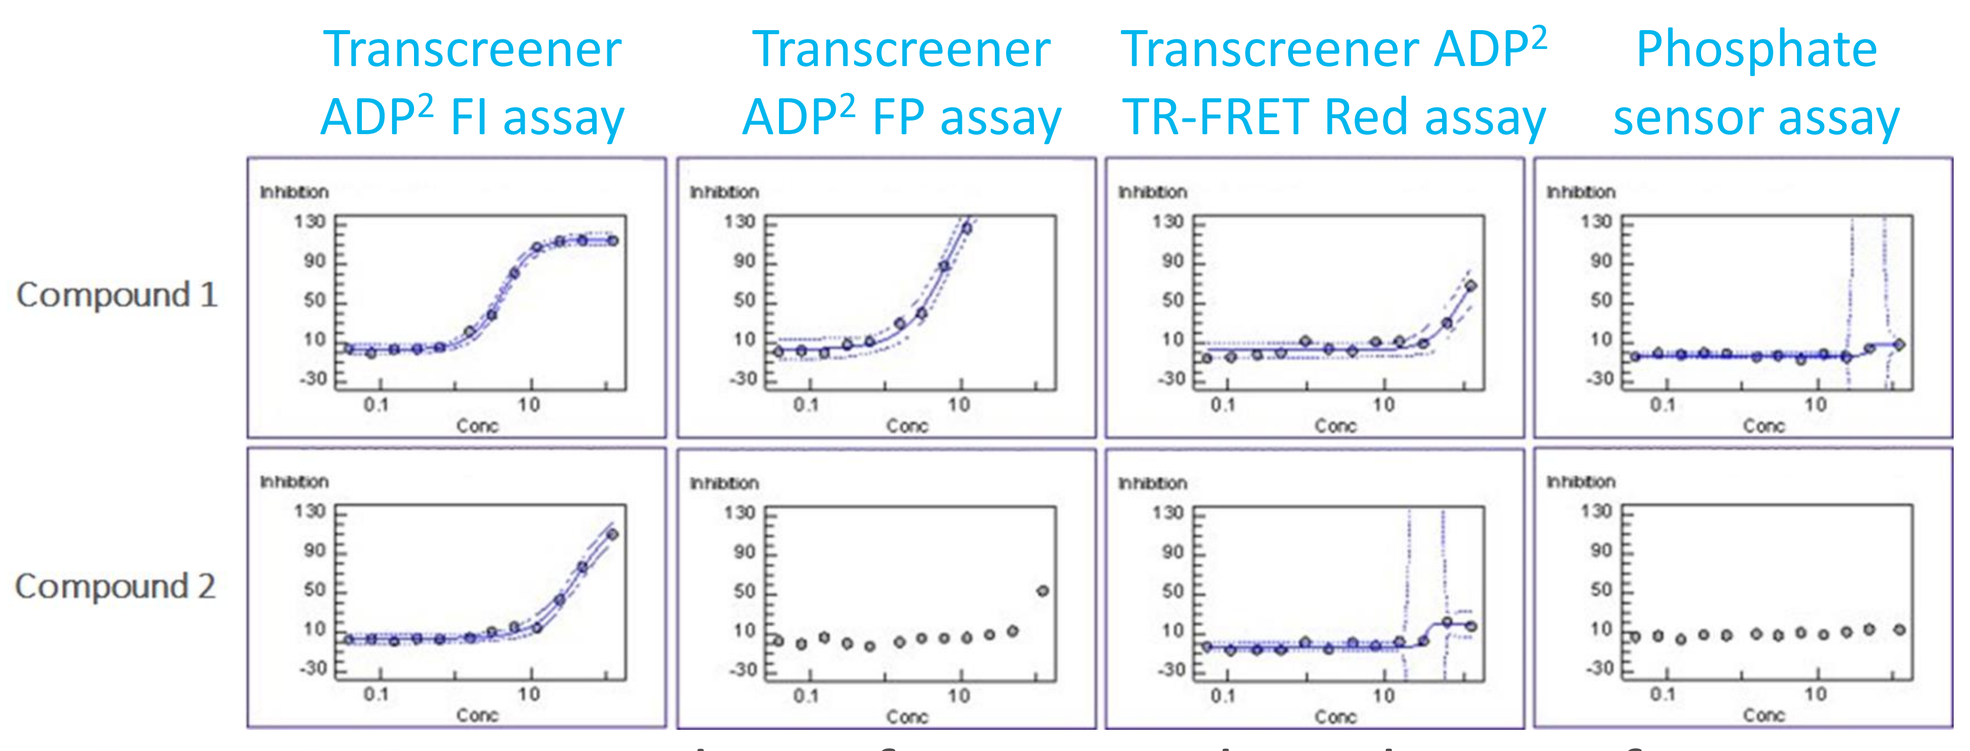

Transcreener ADP² FI assay
Transcreener ADP² FP assay
Transcreener ADP² TR-FRET Red assay
Phosphate sensor assay
Compound 1
Compound 2

Figure 3. Compound interference with each assay format. Interference assays were run in the absence of enzyme by spiking in a set amount of product, any variation from the expected signal in the presence of compound is the result of interference with the detection technology.

The phosphate sensor assay was selected to take forwards.

ASSAY DEVELOPMENT WITH THE PHOSPHATE SENSOR DETECTION REAGENT

Phosphate sensor concentration was optimised to maximise signal to background and reduce cost.

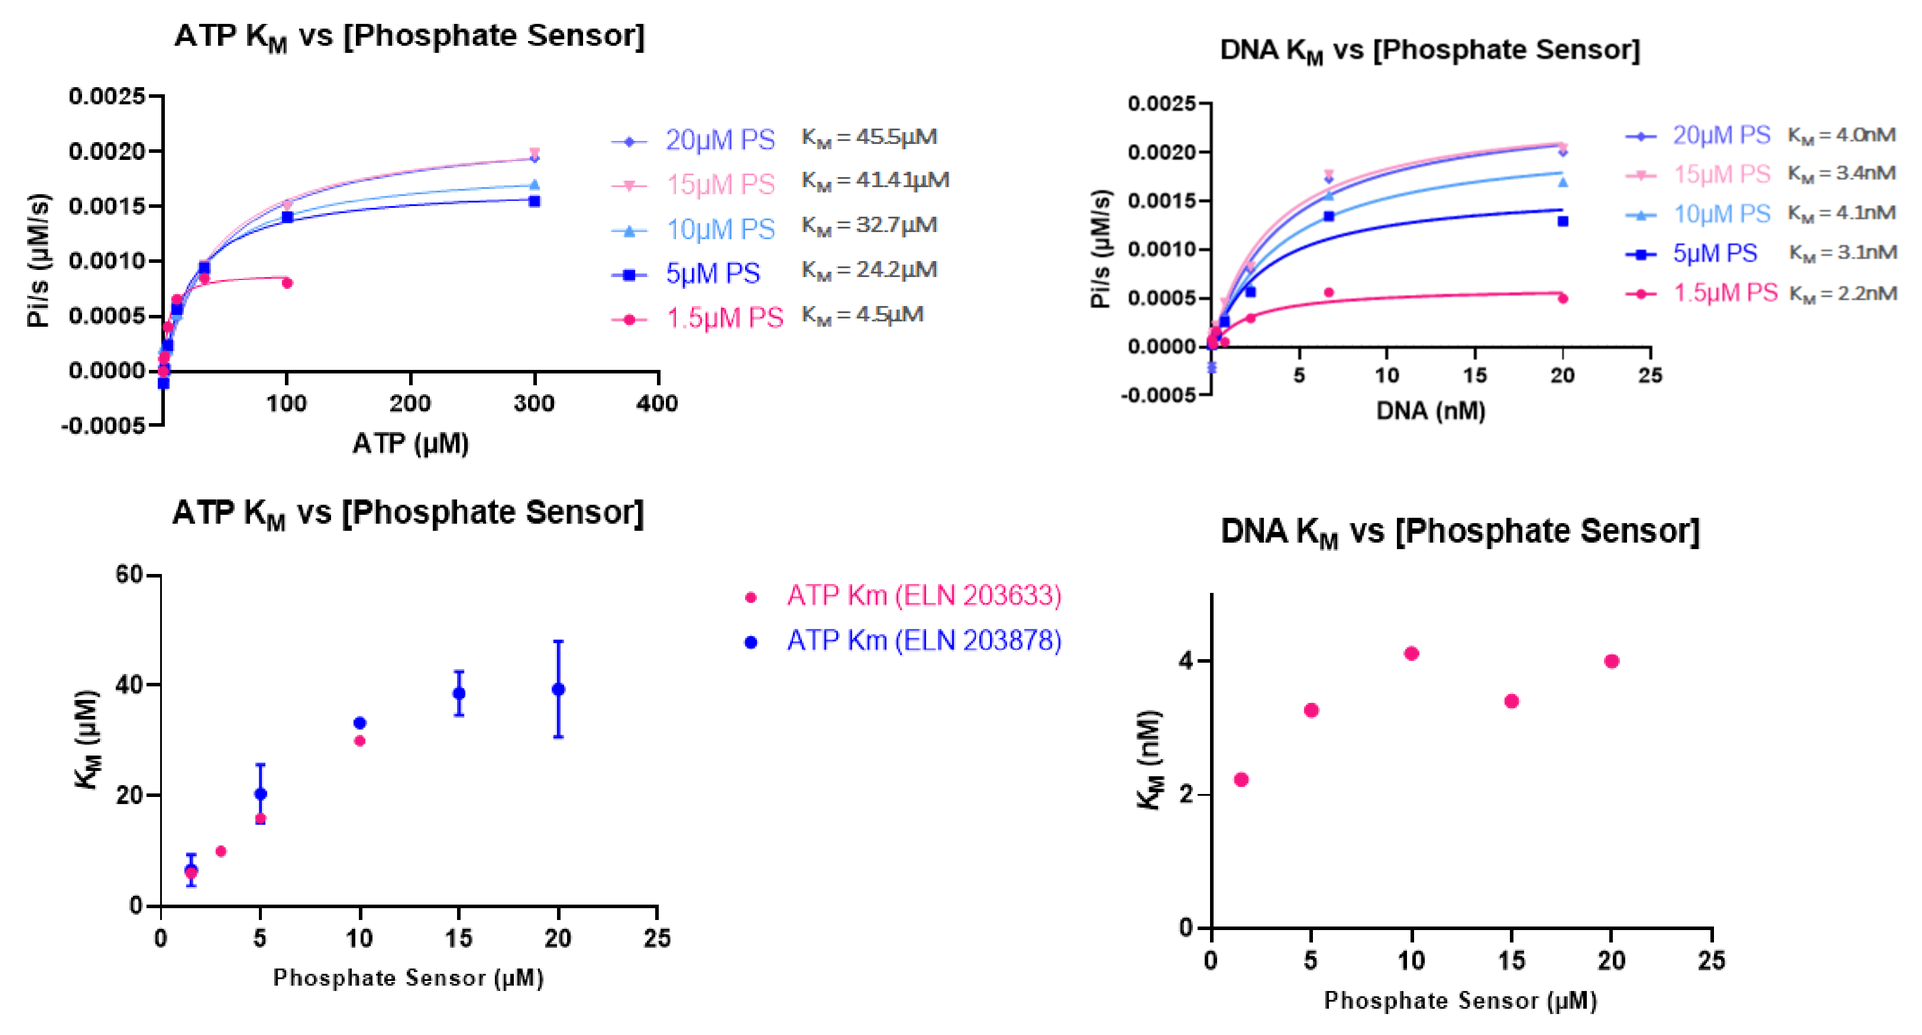

ATP KM vs [Phosphate Sensor]
DNA KM vs [Phosphate Sensor]
20µM PS KM = 45.5µM
15µM PS KM = 41.41µM
10µM PS KM = 32.7µM
5µM PS KM = 24.2µM
1.5µM PS KM = 4.5µM
20µM PS KM = 4.0nM
15µM PS KM = 3.4nM
10µM PS KM = 4.1nM
5µM PS KM = 3.1nM
1.5µM PS KM = 2.2nM
ATP Km (ELN 203633)
ATP Km (ELN 203878)
Pi/s (µM/s)
ATP (µM)
DNA (nM)
KM (µM)
KM (nM)
Phosphate Sensor (µM)

Figure 4. Substrate and detection reagent concentration optimisation. Each substrate was titrated in turn against a range of phosphate sensor concentrations. The second substrate was included in excess to approximate a single substrate system.

Excellent Z’ values were achieved at low enzyme concentrations of 1-4 nM highlighting the sensitivity of this detection reagent.

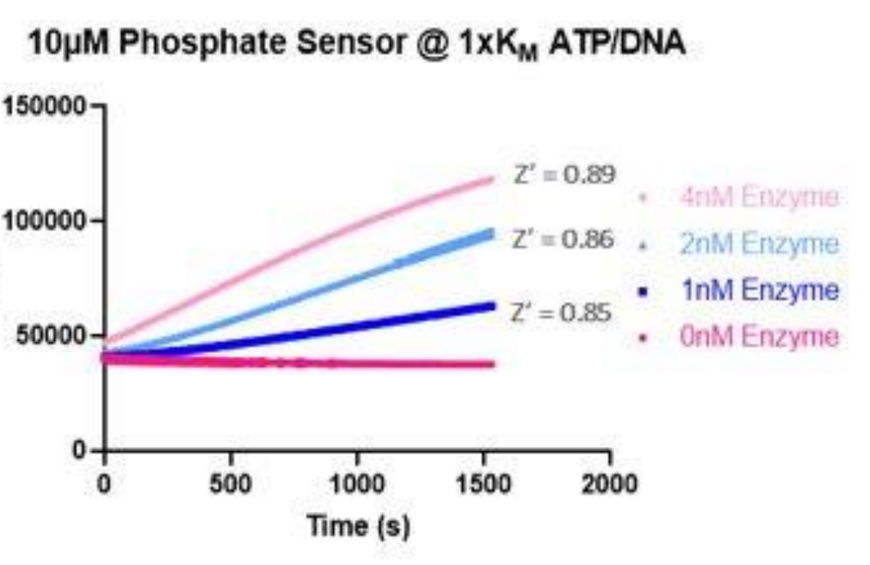

10µM Phosphate Sensor @ 1xKM ATP/DNA
Z' = 0.89
Z' = 0.86
Z' = 0.85
4nM Enzyme
2nM Enzyme
1nM Enzyme
0nM Enzyme
RFU
Time (s)

	IC50 (µM)	pIC50	Graphical result
Inhibitor A	13.997	4.85	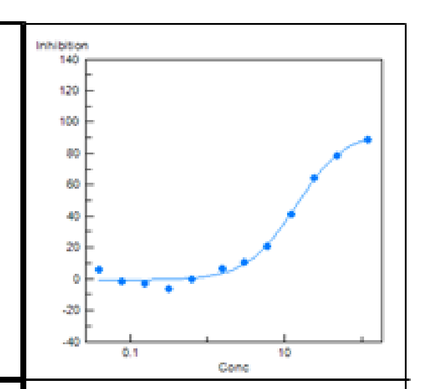
Inhibitor B	2.3625	5.63	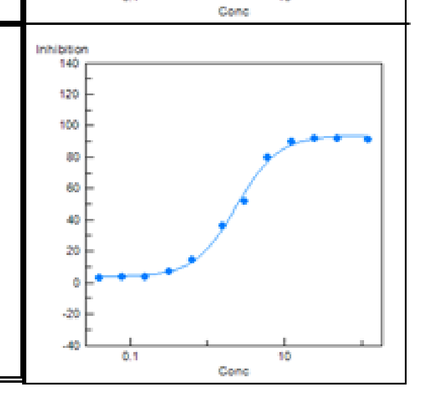

Figure 5. Linearity check and confirmation of compound inhibition under final assay conditions. Compound IC_{50} curves were used to select an appropriate concentration range to use in the MoI assays.

MECHANISM OF INHIBITION DETERMINATION

Detailed mechanism of inhibition (MoI) studies were performed to determine whether compounds were competitive, non-competitive or uncompetitive with respect to the substrates. Kinetic characterisation assays were performed in which K_M and V_{max} kinetic parameters were assessed at different inhibitor concentrations.

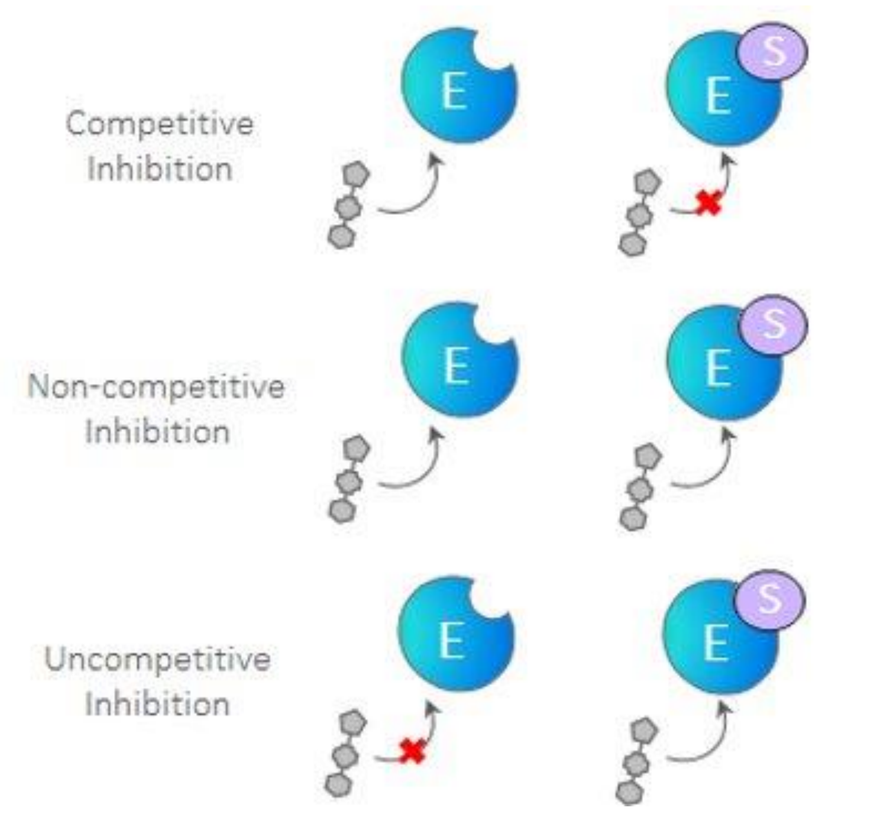

Competitive Inhibition
Non-competitive Inhibition
Uncompetitive Inhibition
E
S

Type of inhibition	K_M	V_{max}	V_{max}/K_M	Binding preference
Competitive	↑	=	↑	E only
Mixed (α > 1)	↑	↓	↓	E > ES
Non-competitive	=	↓	↓	E = ES
Mixed (α < 1)	↓	↓	↓	E < ES
Uncompetitive	↓	↓	=	ES only

Figure 6. Compound inhibition schematics. A competitive compound can only bind to the apo enzyme. A non-competitive compound can bind equally well to the apo enzyme or enzyme complex. An uncompetitive compound can only bind to the enzyme substrate complex. Often the mechanism lies in between one of these cases and is referred to as mixed inhibition.

MECHANISM OF INHIBITION RESULTS

Both compounds were non-competitive with respect to ATP so are able to bind equally well to the apo enzyme or enzyme-ATP complex. The compounds differed in their MoI with respect to the DNA substrate. Compound A was uncompetitive with respect to DNA so is able to only bind to the enzyme-DNA complex. Compound B showed mixed inhibition with respect to DNA so is able to bind to both the apo enzyme and enzyme-DNA complex with a preference for the apo enzyme.

Inhibitor A

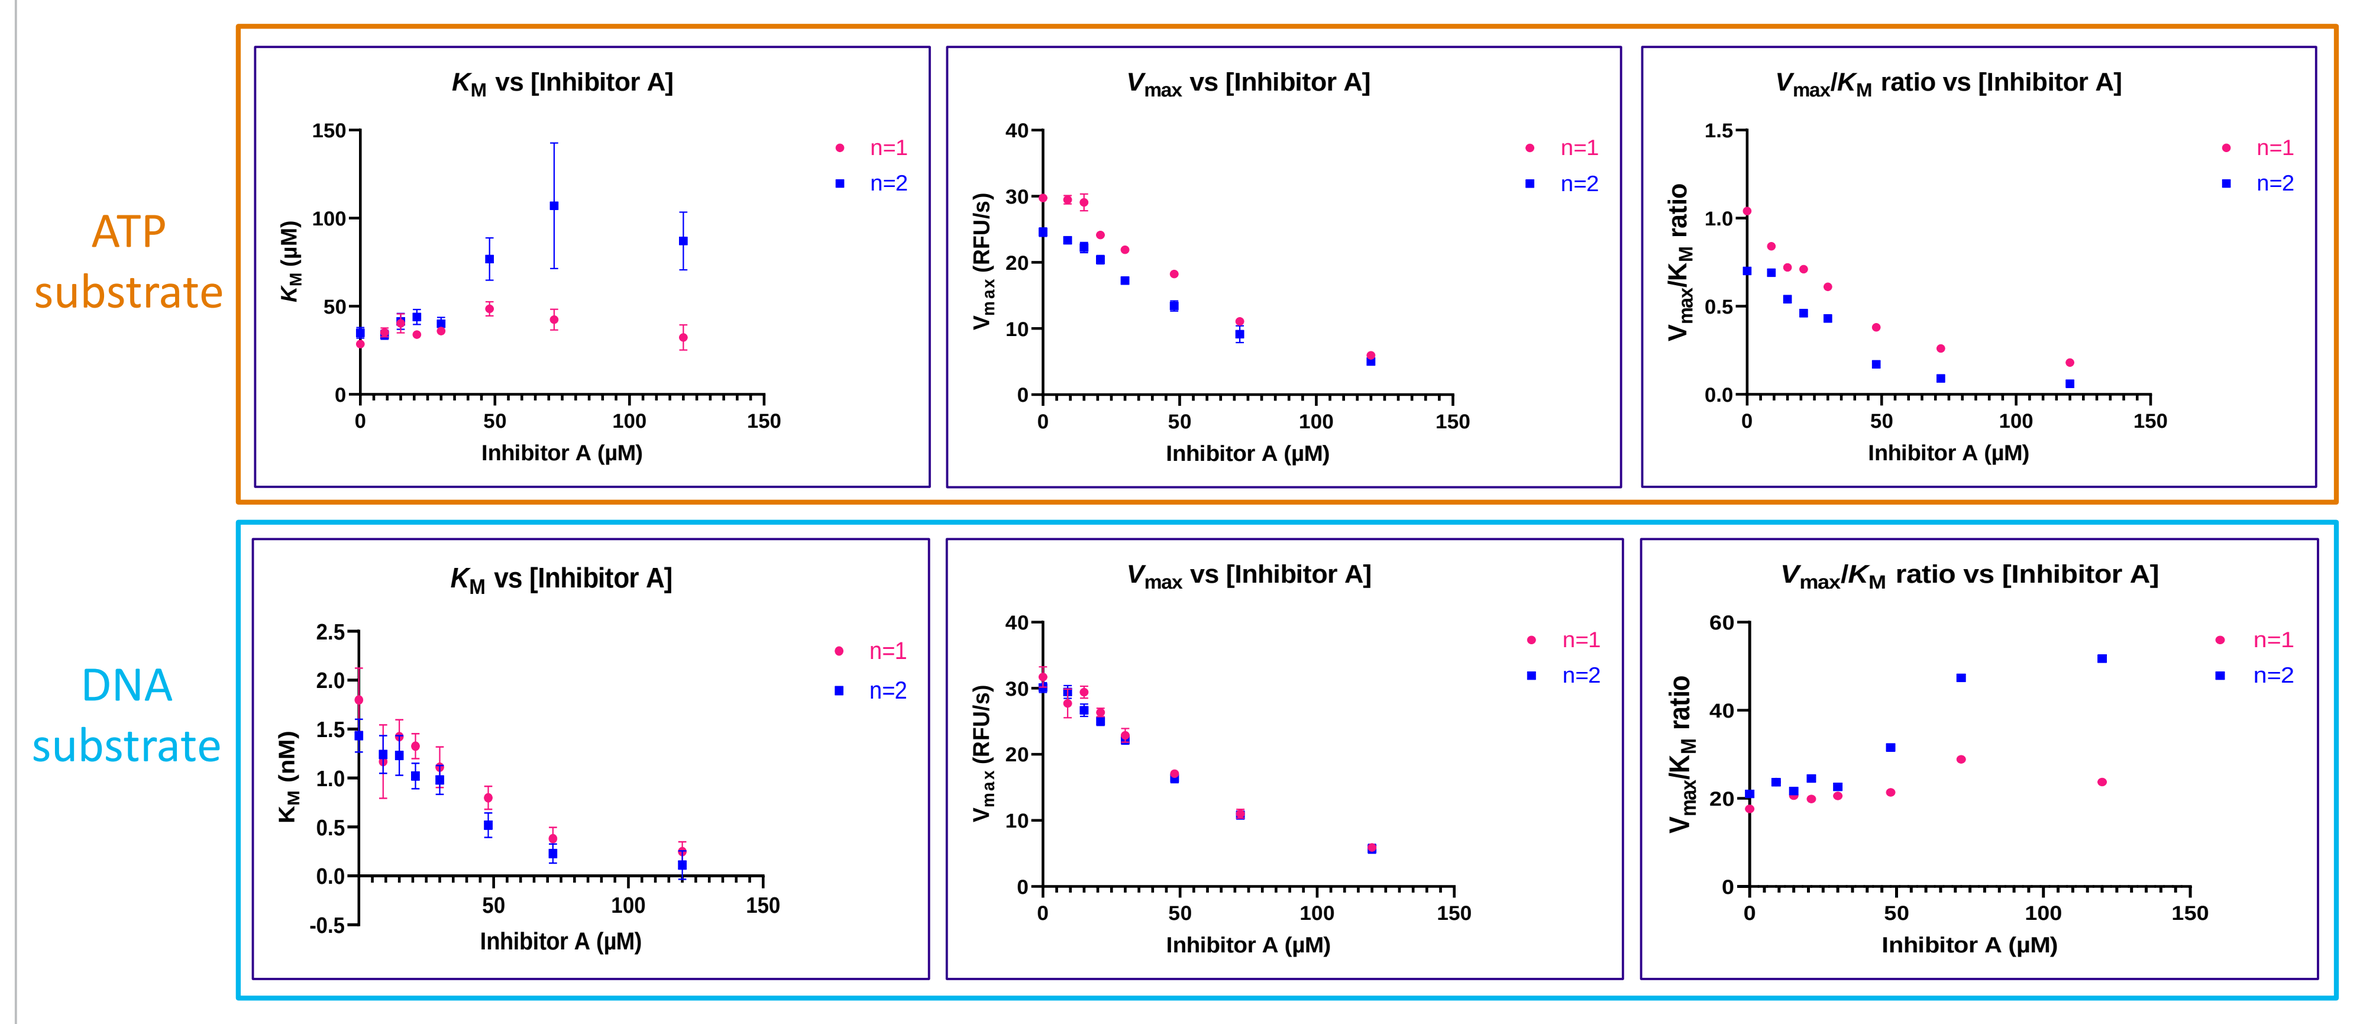

ATP substrate
DNA substrate
KM vs [Inhibitor A]
Vmax vs [Inhibitor A]
Vmax/KM ratio vs [Inhibitor A]
n=1
n=2
Inhibitor A (µM)

Inhibitor B

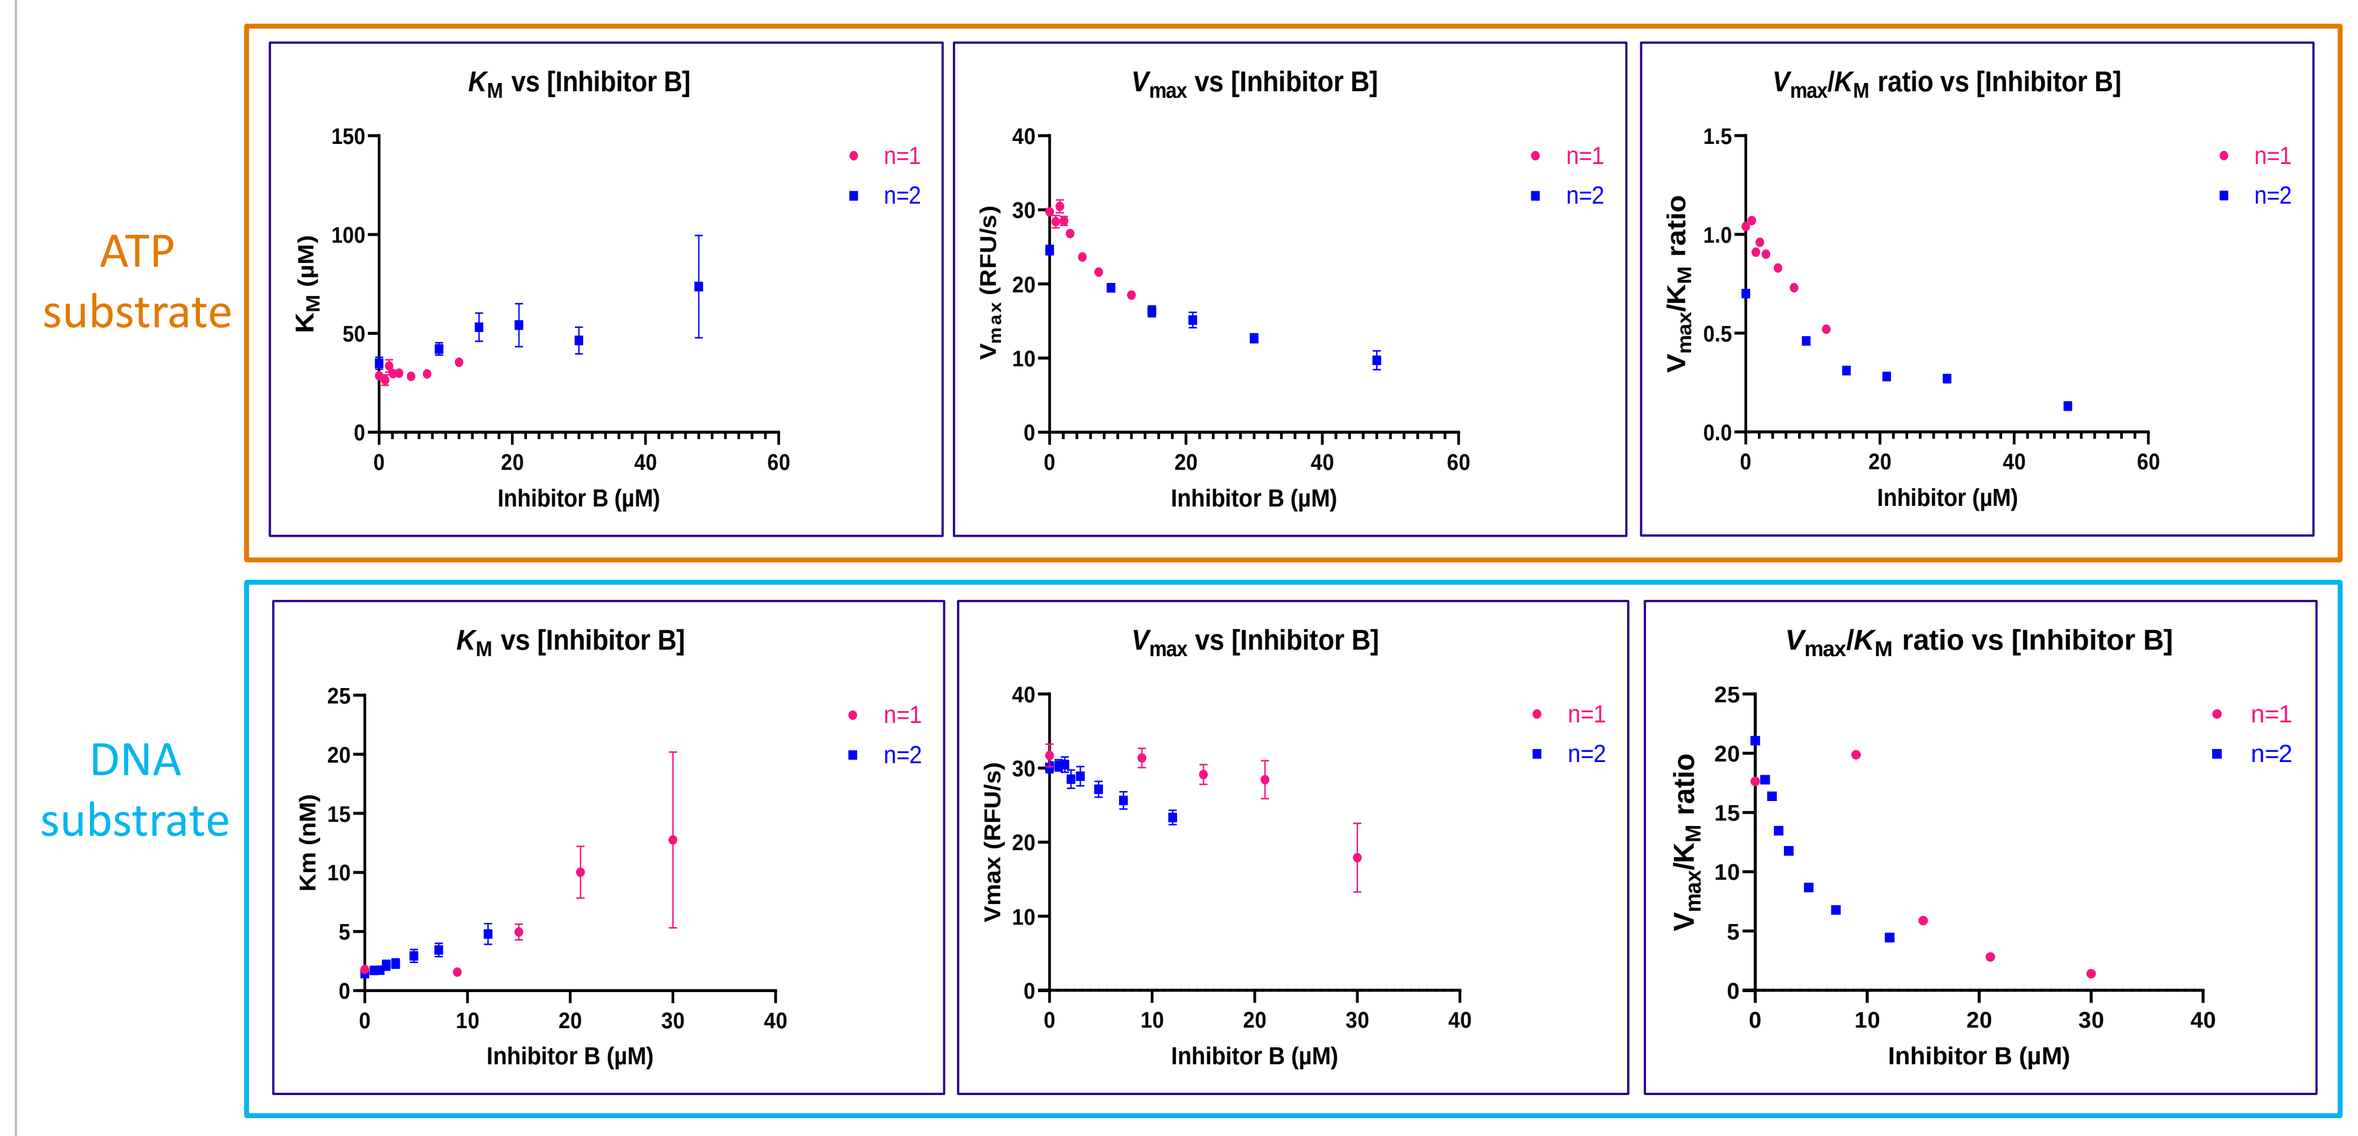

ATP substrate
DNA substrate
KM vs [Inhibitor B]
Vmax vs [Inhibitor B]
Vmax/KM ratio vs [Inhibitor B]
n=1
n=2
Inhibitor B (µM)

Figure 7. Detailed MoI studies with two compounds of interest. Initial rates were calculated in the presence of 10 µM phosphate sensor reagent between 5-16 minutes. Initial rates in RFU/second were plotted against substrate concentration. Using GraphPad Prism this data was fitted to equations for competitive, non-competitive, uncompetitive and mixed inhibition and the best fit determined using AIC. This data was also fitted to the Michaelis Menten equation to determine the K_M and V_{max} values plotted here to show the trend with increasing compound concentration which can indicate the type of inhibition. The second substrate was included in excess to approximate a single substrate system.

SUMMARY

Several detection technologies for continuous measurement of ATPase activity were assessed here. The phosphate sensor reagent was selected as the most promising as it was not prone to interference with our compounds and was very sensitive. We successfully used this assay to perform detailed MoI studies and were able to demonstrate differences in the MoI of representative compounds from two series of interest. Using the kinetic assay format for these studies significantly reduced both resource and reagent consumption compared to using an endpoint assay format.

CANCER RESEARCH UK | THERAPEUTIC DISCOVERY LABORATORIES